Systems Pharmacology Model Development to Provide Physiologically-Based Interpretation and Drug Development Decision Support in Osteoporosis and Other Bone-Related Diseases

Matthew M. Riggs, Ph.D.

Metrum Research Group LLC; 2 Tunxis Road Suite 112; Tariffville, CT USA

METRUM RESEARCH GROUP

ACKNOWLEDGEMENTS

The work reviewed in this poster has evolved with the dedicated efforts of many contributors, as cited, including (alphabetically): *Kyle T. Baron, Ph.D.*[1], *Marc R. Gastonguay, Ph.D.*[1], *Steve Martin, Ph.D.*[2], *Alanna Ocampo-Pelland, M.S.,Ph.D. Student*[1], *Mark C. Peterson, Ph.D.*[2,3], *Elodie Plan, Ph.D.*[2,4], *Piet van der Graaf, Ph.D.*[2]
[1]**Metrum Research Group LLC,** [2]**Pfizer Inc,** [3]**formerly at Amgen Inc,** [4]**currently at Uppsala University**

ABSTRACT

Models describing PK-PD, disease progression, and systems biology provide the components for a **Multiscale Systems Pharmacology Model (MSPM)** with a goal to enable in silico exploration of the ramifications of system perturbations, e.g., those caused by disease, genetic variation, and therapeutic intervention(s). In doing so, the MSPM approach offers a continuum for translating experimental data with clinical biomarkers and outcomes allowing researchers to probe target pathways, understand potential patient-to-patient sensitivities, and ultimately design more efficient and informative clinical trials. Applied broadly, these models instill rapid, seamless knowledge transfer across the research scale by enabling colleagues from multiple disciplines to communicate with a common set of expectations and understanding of these oftentimes complex systems. MSPMs can thereby serve as repositories that contain and connect data, experimentations, overall comprehension, and assumptions of the system.

A MSPM describing bone mineral homeostasis and bone remodeling will be used as an example of how to develop and apply such a model with a focus on MSPM extensibility to evolving and broadening research goals. A brief history of the model construction will be provided with its initial focus on osteoporosis treatment[1] and continued expansion to other natural progressions and diseases involving bone, e.g., natural estrogen loss during menopause transition[2] and during endometriosis treatment,[3] as well as Chronic Kidney Disease-Mineral and Bone Disorder.[4] Additionally, a "middle-out"[5] approach to ongoing research efforts include the translation of bone marker effects to BMD prediction and subsequent fracture risk[8] and inclusion of Vitamin D kinetics. An overview and update on this research, as well as future plans, will be provided.

INTRODUCTION – MODEL BACKGROUND

Multiscale Systems Biology / Pharmacology Models (*Figure 1*)

- Biologic systems expressed as mathematical expressions
- Quantify timecourses, magnitudes of changes (e.g., natural decays, interactions)
- Serve as in silico probes of biologic perturbation (e.g., disease, genetic variation)
- Multiscale systems pharmacology model (**MSPM**): include pharmacologic effects

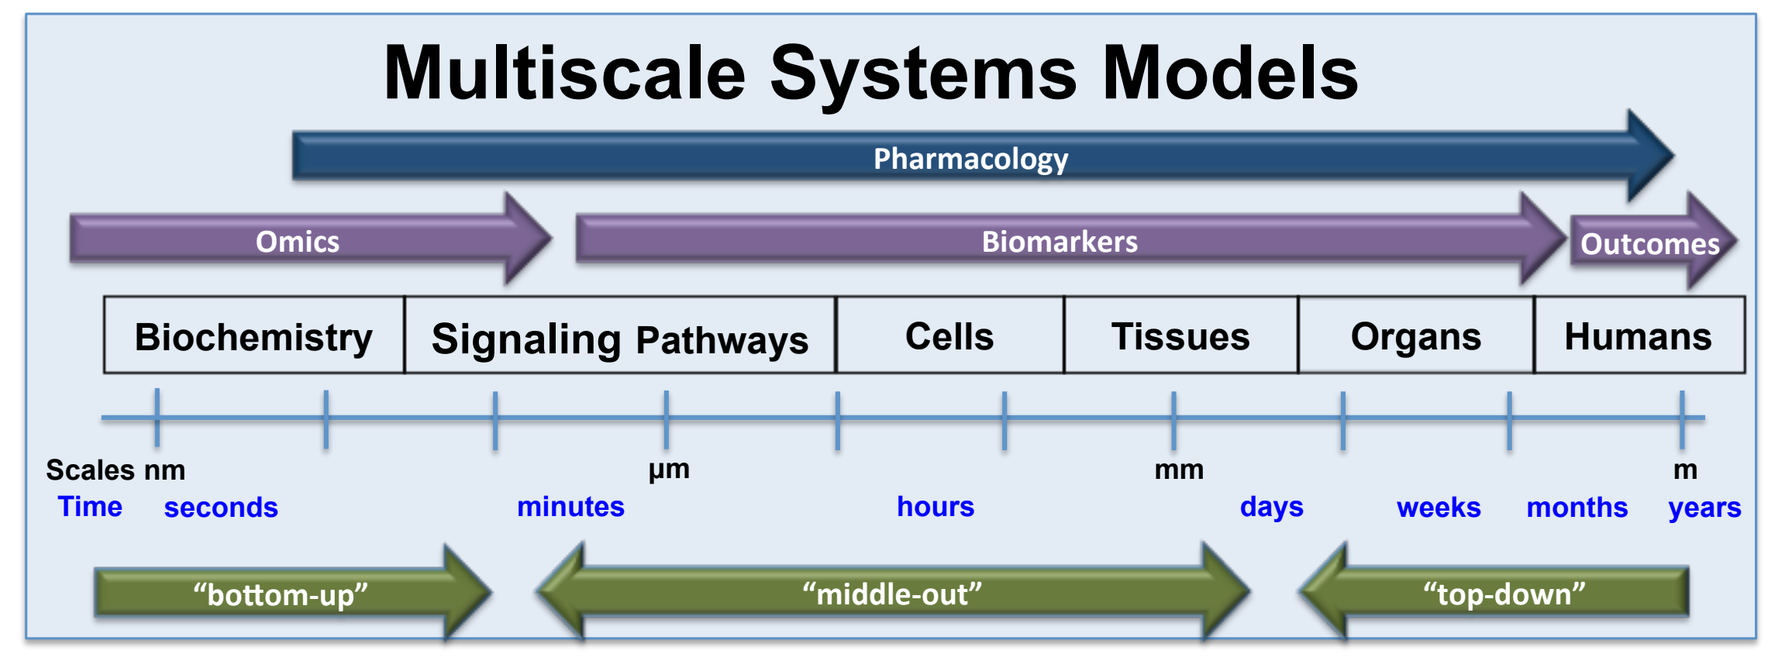
Multiscale Systems Models
Pharmacology
Omics
Biomarkers
Outcomes
Biochemistry
Signaling Pathways
Cells
Tissues
Organs
Humans
Scales nm
μm
mm
m
Time seconds
minutes
hours
days
weeks
months
years
"bottom-up"
"middle-out"
"top-down"

Figure 1: *Defining multiscale systems models and terminology; reproduced from Riggs 2011*[5]

MSPM of Bone Mineral Homeostasis and Remodeling (*Figures 2, 3*)

- Mathematical (differential equations) construct from experimental and clinical data
- Scales: Cell signaling → organ functions → bone turnover markers (BTMs) → BMD
- Applications:
 - **Denosumab: PTH, serum calcium, BTMs,[1] and lumbar spine BMD[6, 7]**
 - Teriparatide: PTH, serum calcium, and BTMs[1]
 - Disease/Aging [CKD-MBD,[4] **menopause and endometriosis**[3]]: BTMs, BMD and fracture risk[8]
- Software: R (www.R-project.org/)[9]
- Model code available through:
 - Original publication[1] (open-source): www.opendiseasemodels.org
 - GnRH and menopause[3]: See CPT:PSP online supplemental material

Example I: Denosumab (RANK-L Inhibition) (*Figure 4*) [6, 7]

- **MSPM Qualification:** Can the MSPM predict clinical observations that were not included in the original model development? In this case, endpoint data collected from a separate clinical study

Example II: Menopause and GnRH Modulation (*Figure 5*)

- Provide model-based decision support for gonadotropin releasing hormone (GnRH) modulator programs intended for the management of EM

Example III: Link BMD Change with Fracture Risk (*Figure 9*)

- Develop model simultaneously characterizing BMD and fracture risk

INTRODUCTION – MULTISCALE SYSTEMS PHARMACOLOGY MODEL OVERVIEW

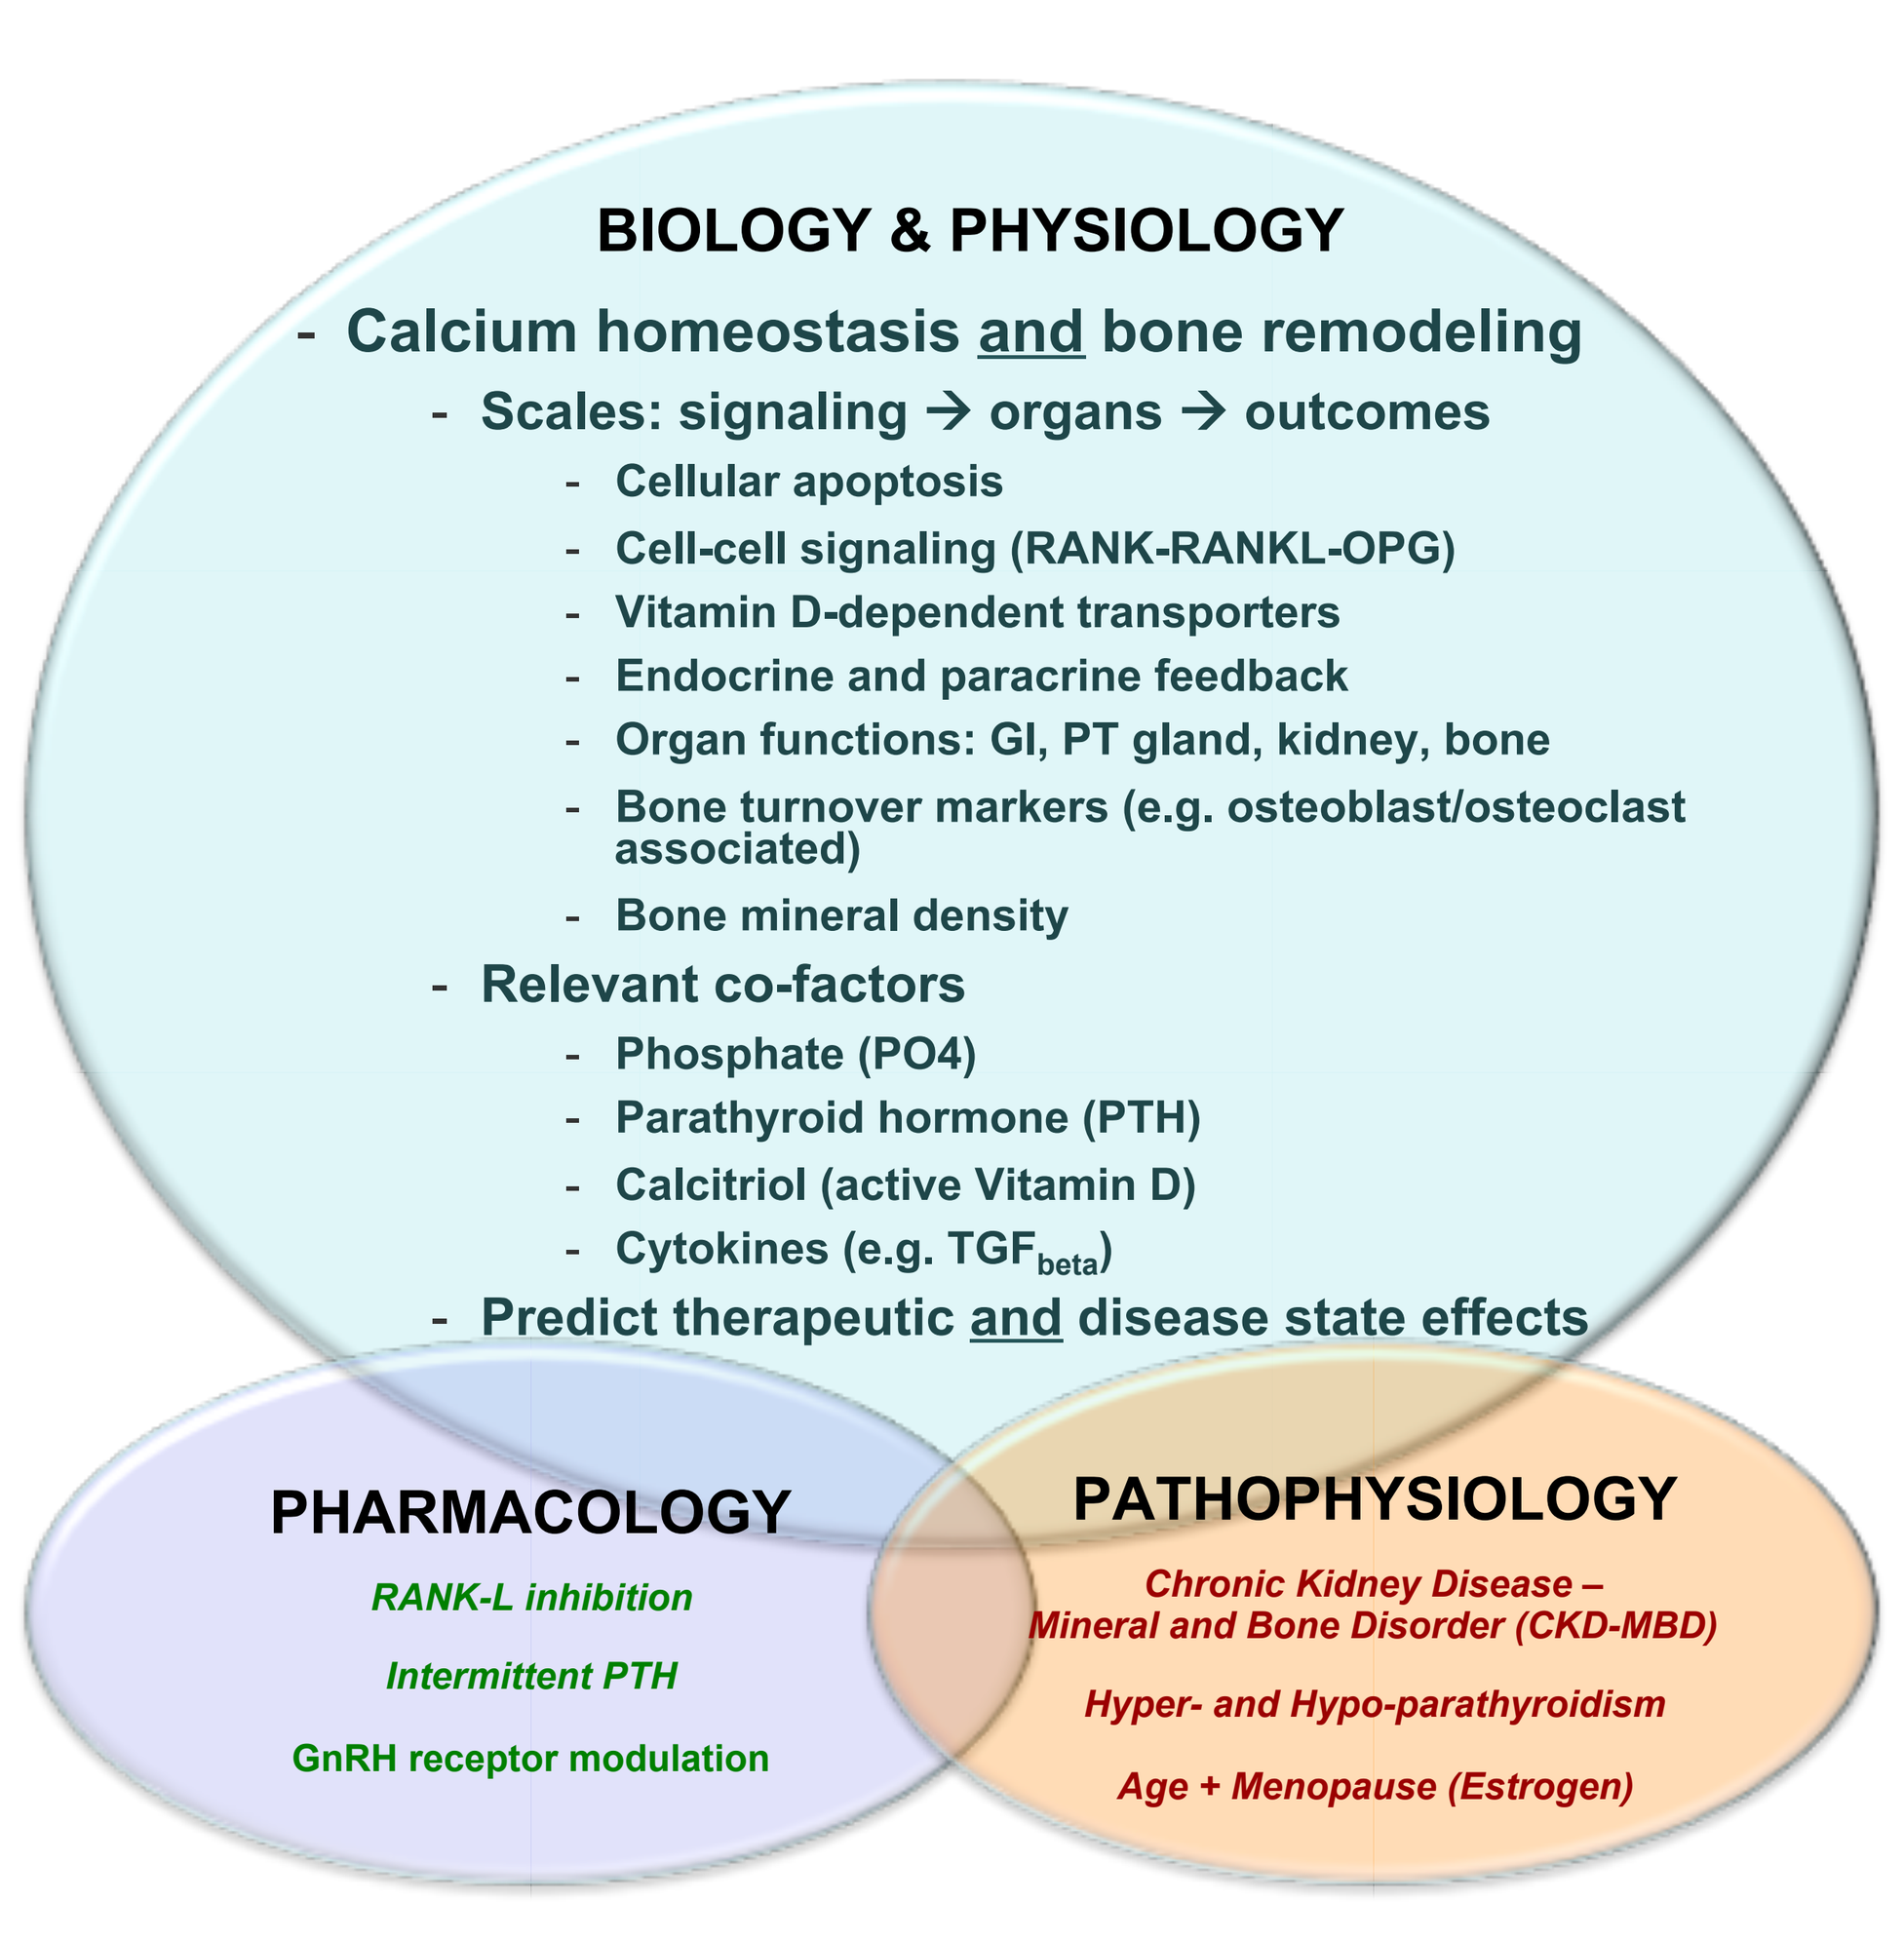
BIOLOGY & PHYSIOLOGY
- Calcium homeostasis and bone remodeling
- Scales: signaling → organs → outcomes
- Cellular apoptosis
- Cell-cell signaling (RANK-RANKL-OPG)
- Vitamin D-dependent transporters
- Endocrine and paracrine feedback
- Organ functions: GI, PT gland, kidney, bone
- Bone turnover markers (e.g. osteoblast/osteoclast associated)
- Bone mineral density
- Relevant co-factors
- Phosphate (PO4)
- Parathyroid hormone (PTH)
- Calcitriol (active Vitamin D)
- Cytokines (e.g. TGFbeta)
- Predict therapeutic and disease state effects
PHARMACOLOGY
RANK-L inhibition
Intermittent PTH
GnRH receptor modulation
PATHOPHYSIOLOGY
Chronic Kidney Disease – Mineral and Bone Disorder (CKD-MBD)
Hyper- and Hypo-parathyroidism
Age + Menopause (Estrogen)

Figure 2: *MSPM conceptual framework and content overview.*

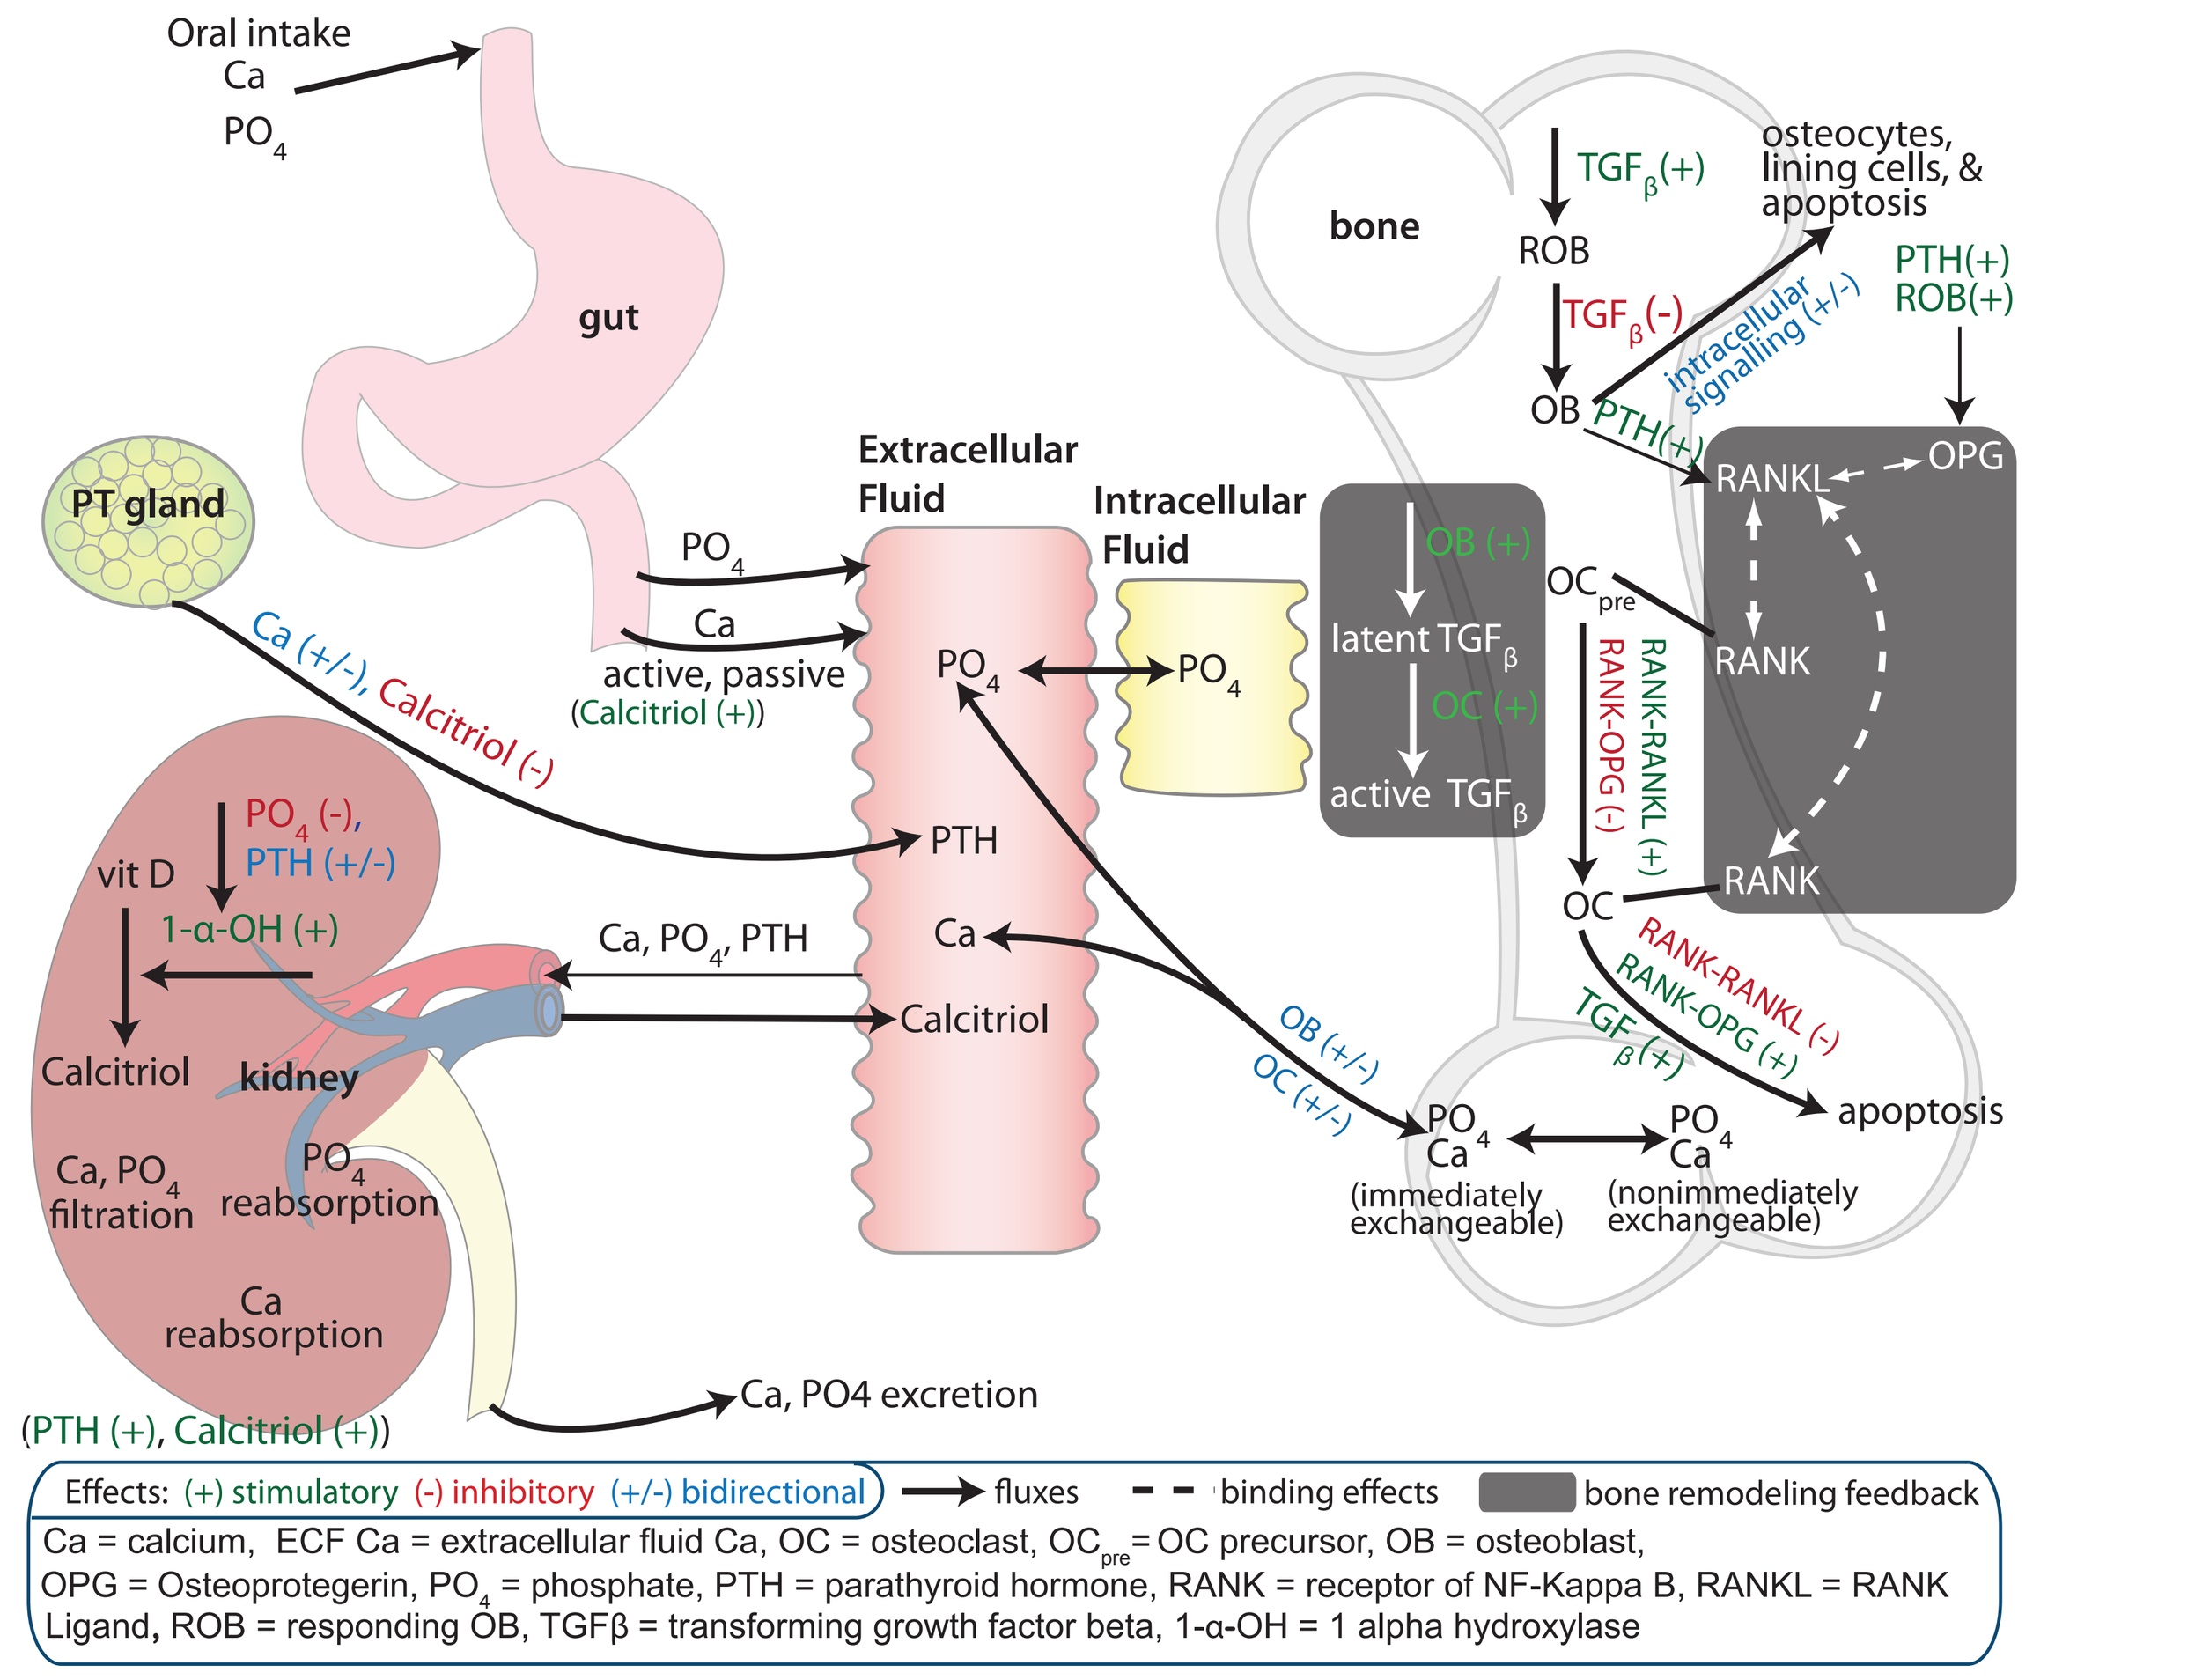
Oral intake
Ca
PO4
gut
PT gland
Ca (+/-), Calcitriol (-)
PO4
Ca
active, passive
(Calcitriol (+))
Extracellular Fluid
Intracellular Fluid
PO4
PTH
Ca
Calcitriol
vit D
PO4 (-), PTH (+/-)
1-α-OH (+)
Calcitriol
kidney
Ca, PO4 filtration
PO4 reabsorption
Ca reabsorption
Ca, PO4, PTH
(PTH (+), Calcitriol (+))
Ca, PO4 excretion
bone
TGFβ(+)
ROB
TGFβ(-)
OB
PTH(+)
intracellular signalling (+/-)
osteocytes, lining cells, & apoptosis
PTH(+)
ROB(+)
latent TGFβ
OB (+)
OC (+)
active TGFβ
OCpre
RANK-RANKL (+)
RANK-OPG (-)
RANKL
OPG
RANK
OC
RANK-RANKL (-)
RANK-OPG (+)
TGFβ (+)
apoptosis
OB (+/-)
OC (+/-)
PO4 Ca (immediately exchangeable)
PO4 Ca (nonimmediately exchangeable)
Effects: (+) stimulatory (-) inhibitory (+/-) bidirectional
fluxes
binding effects
bone remodeling feedback
Ca = calcium, ECF Ca = extracellular fluid Ca, OC = osteoclast, OCpre = OC precursor, OB = osteoblast,
OPG = Osteoprotegerin, PO4 = phosphate, PTH = parathyroid hormone, RANK = receptor of NF-Kappa B, RANKL = RANK Ligand, ROB = responding OB, TGFβ = transforming growth factor beta, 1-α-OH = 1 alpha hydroxylase

Figure 3: *Schematic of physiologically-based, multiscale systems pharmacology model; modified from Figure 1 of Peterson and Riggs, 2010.* [1]

METHODS – EXAMPLE I: QUALIFYING PREDICTIONS OF DENOSUMAB EFFICACY

The Receptor Activator of Nuclear Factor-κ B (RANK)-RANK Ligand (RANKL)-Osteoprotegerin (OPG) system

$\Uparrow$ OC differentiation and $\Uparrow$ OC activation: RANK–RANKL

$\Downarrow$ OC differentiation and $\Uparrow$ OC apoptosis: RANKL–OPG, RANKL–denosumab

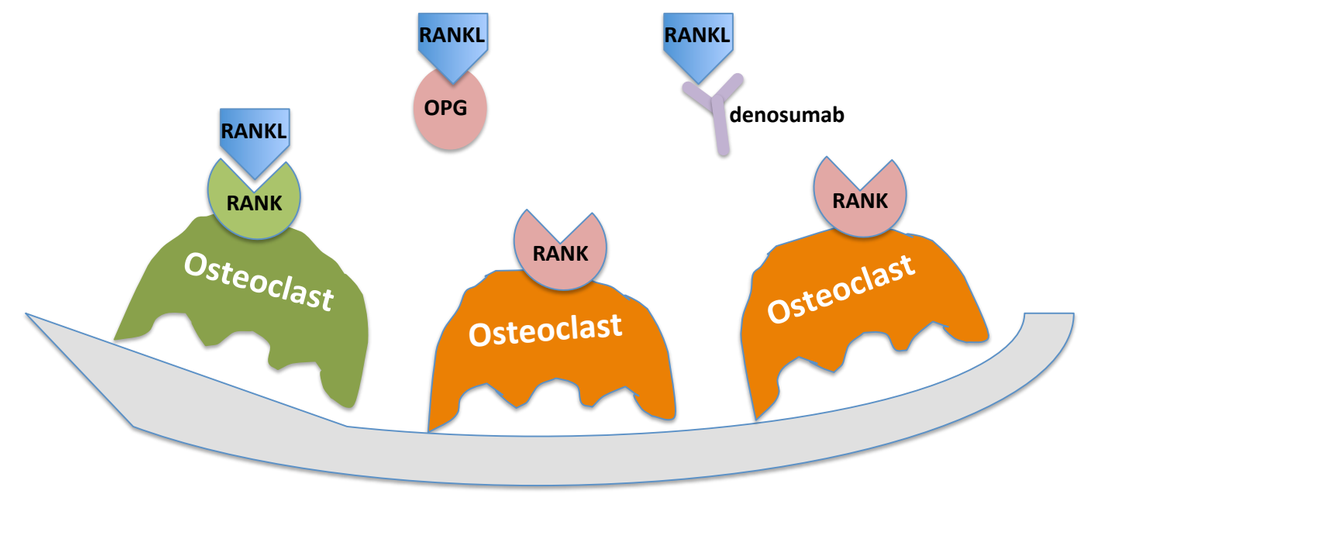
RANKL
OPG
RANKL
denosumab
RANKL
RANK
RANK
RANK
Osteoclast
Osteoclast
Osteoclast

Figure 4: *Depiction of RANK-RANKL-OPG and denosumab interaction.*

Denosumab[10]
- Fully human monoclonal antibody
- Binds to RANKL with high affinity and specificity
- Blocks interaction of RANKL with RANK
- Mimics endogenous effects of OPG

Denosumab–RANKL binding

- ↓ available RANKL
- ↓ RANK–RANKL interaction
- ↓ Osteoclast activity (serum C-telopeptide, CTx)
- ↓ Activation of TGFβ
- ↓ Osteoblast activity (bone-specific alkaline phosphatase, BSAP)
- ↑ bone mineral density (BMD), expressed mathematically[6] as:

$$\frac{d(LS\ BMD)}{dt} = k_{in,BMD} \cdot \left(\frac{\mathrm{BSAP}}{\mathrm{BSAP_{baseline}}}\right)^{\gamma_{OB}} - k_{out,BMD} \cdot \left(\frac{\mathrm{CT_x}}{\mathrm{CT_{x_{baseline}}}}\right)^{\gamma_{OC}} \cdot LS\ BMD$$

$$\gamma_{OB} = 0.0793 \quad \gamma_{OC} = 0.0679 \quad k_{\mathrm{out,BMD}} = 0.000145\ \mathrm{h}^{-1}$$

EXAMPLE II: A BALANCED SAFETY / EFFICACY TARGET FOR ENDOMETRIOSIS

Gonadotropin Releasing Hormone (GnRH) Modulation[3]

- Estrogen (E2)-mediated effects on bone turnover modeled through actions on TGF-β and RANKL using data from menopause transition[2, 3]

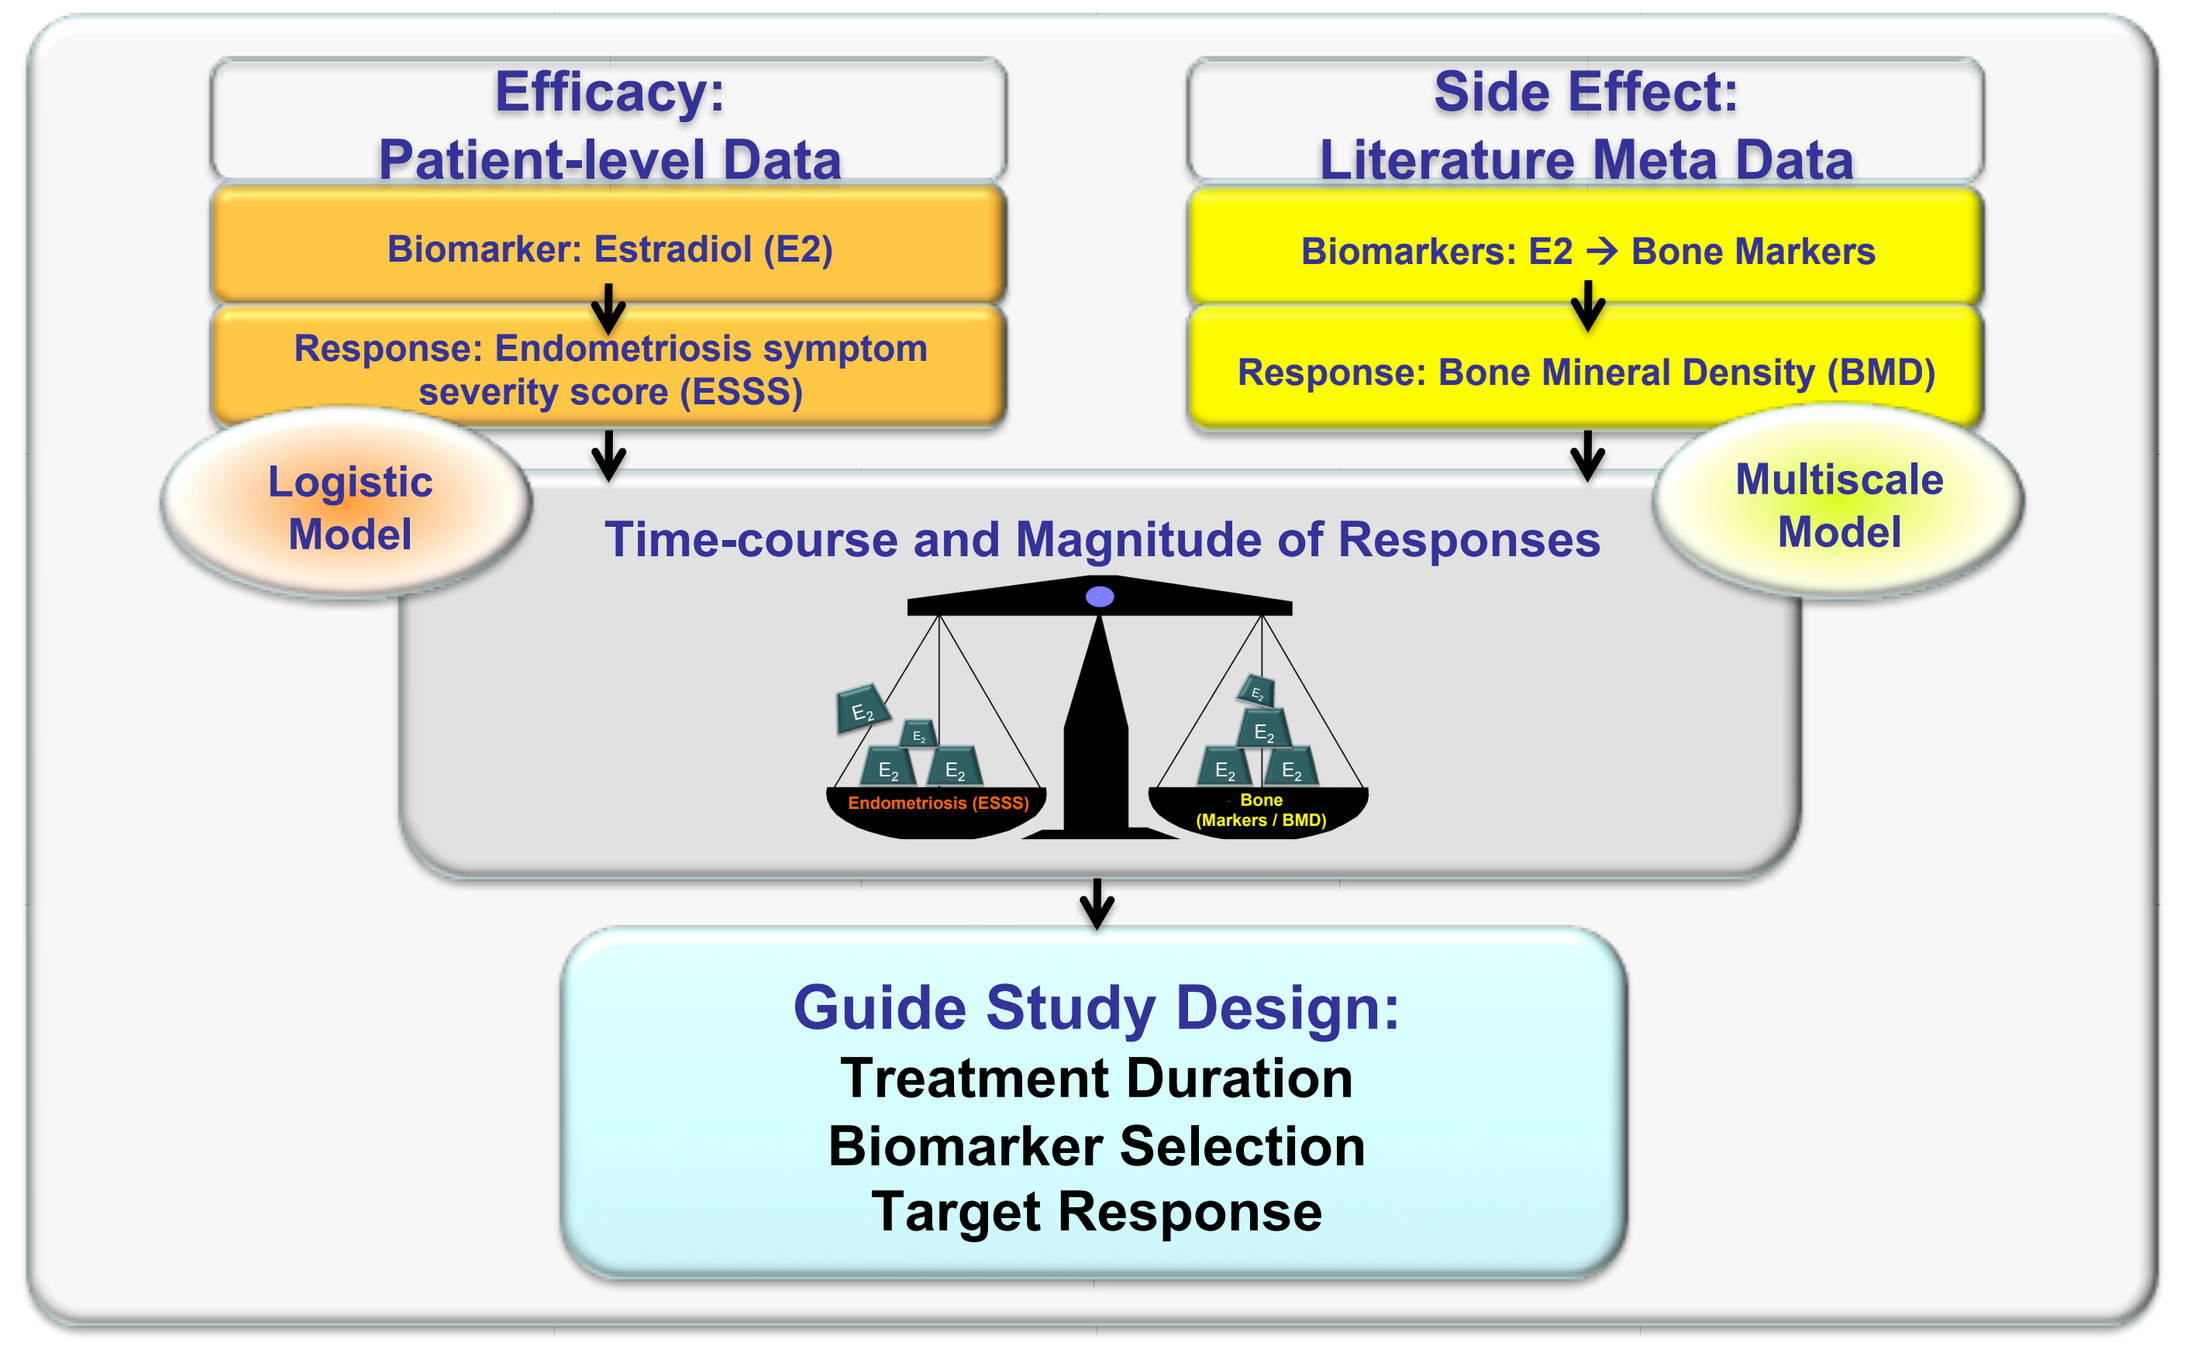
Efficacy: Patient-level Data
Biomarker: Estradiol (E2)
Response: Endometriosis symptom severity score (ESSS)
Side Effect: Literature Meta Data
Biomarkers: E2 → Bone Markers
Response: Bone Mineral Density (BMD)
Logistic Model
Multiscale Model
Time-course and Magnitude of Responses
Endometriosis (ESSS)
Bone (Markers / BMD)
Guide Study Design:
Treatment Duration
Biomarker Selection
Target Response

Figure 5: *Quantifying the balance between efficacy (endometriosis symptom relief) and safety (loss of BMD) .*

- MSPM describes E2 → bone marker → BMD relationship
- Modeled relationship between GnRH suppression and E2
- Evaluated E2, bone maker, BMD effects sizes and time-courses

METHODS – EXAMPLE III: EXTENTION TO INCLUDE FRACTURE RISK

Subject Level Data: 2005-2008 NHANES[11]

- Demographics, dual energy X-ray absorptiometry, body measures, osteoporosis, and reproductive health datasets
- 1605 postmenopausal ♀ of 63 yr mean age and 45 yr mean final menstrual period (FMP) age; 1 femoral neck BMD measure and 0–5 (204 total) fracture events each

Repeated Time-to-Event Fracture Risk Model[12]

- **Exponential survival (fraction without fracture) time:** $S(t) = e^{-(\int_{t_{j-1}}^{t_j} h(u)du)^{\alpha}}$
- **Hazard: time-dependence through BMD(t):** $h(t) = e^{\theta_h \times (1+\theta_{BMD} \times (\mathrm{BMD}(t) - \overline{\mathrm{BMD}}))}$
- BMI, ethnicity, and FMP_{age} included as covariates on BMD model

RESULTS – MSPM QUALIFICATION USING *FREEDOM* DATA (EXAMPLE I)

MSPM Predicted Lumbar Spine BMD and Bone Marker Responses[6, 7]

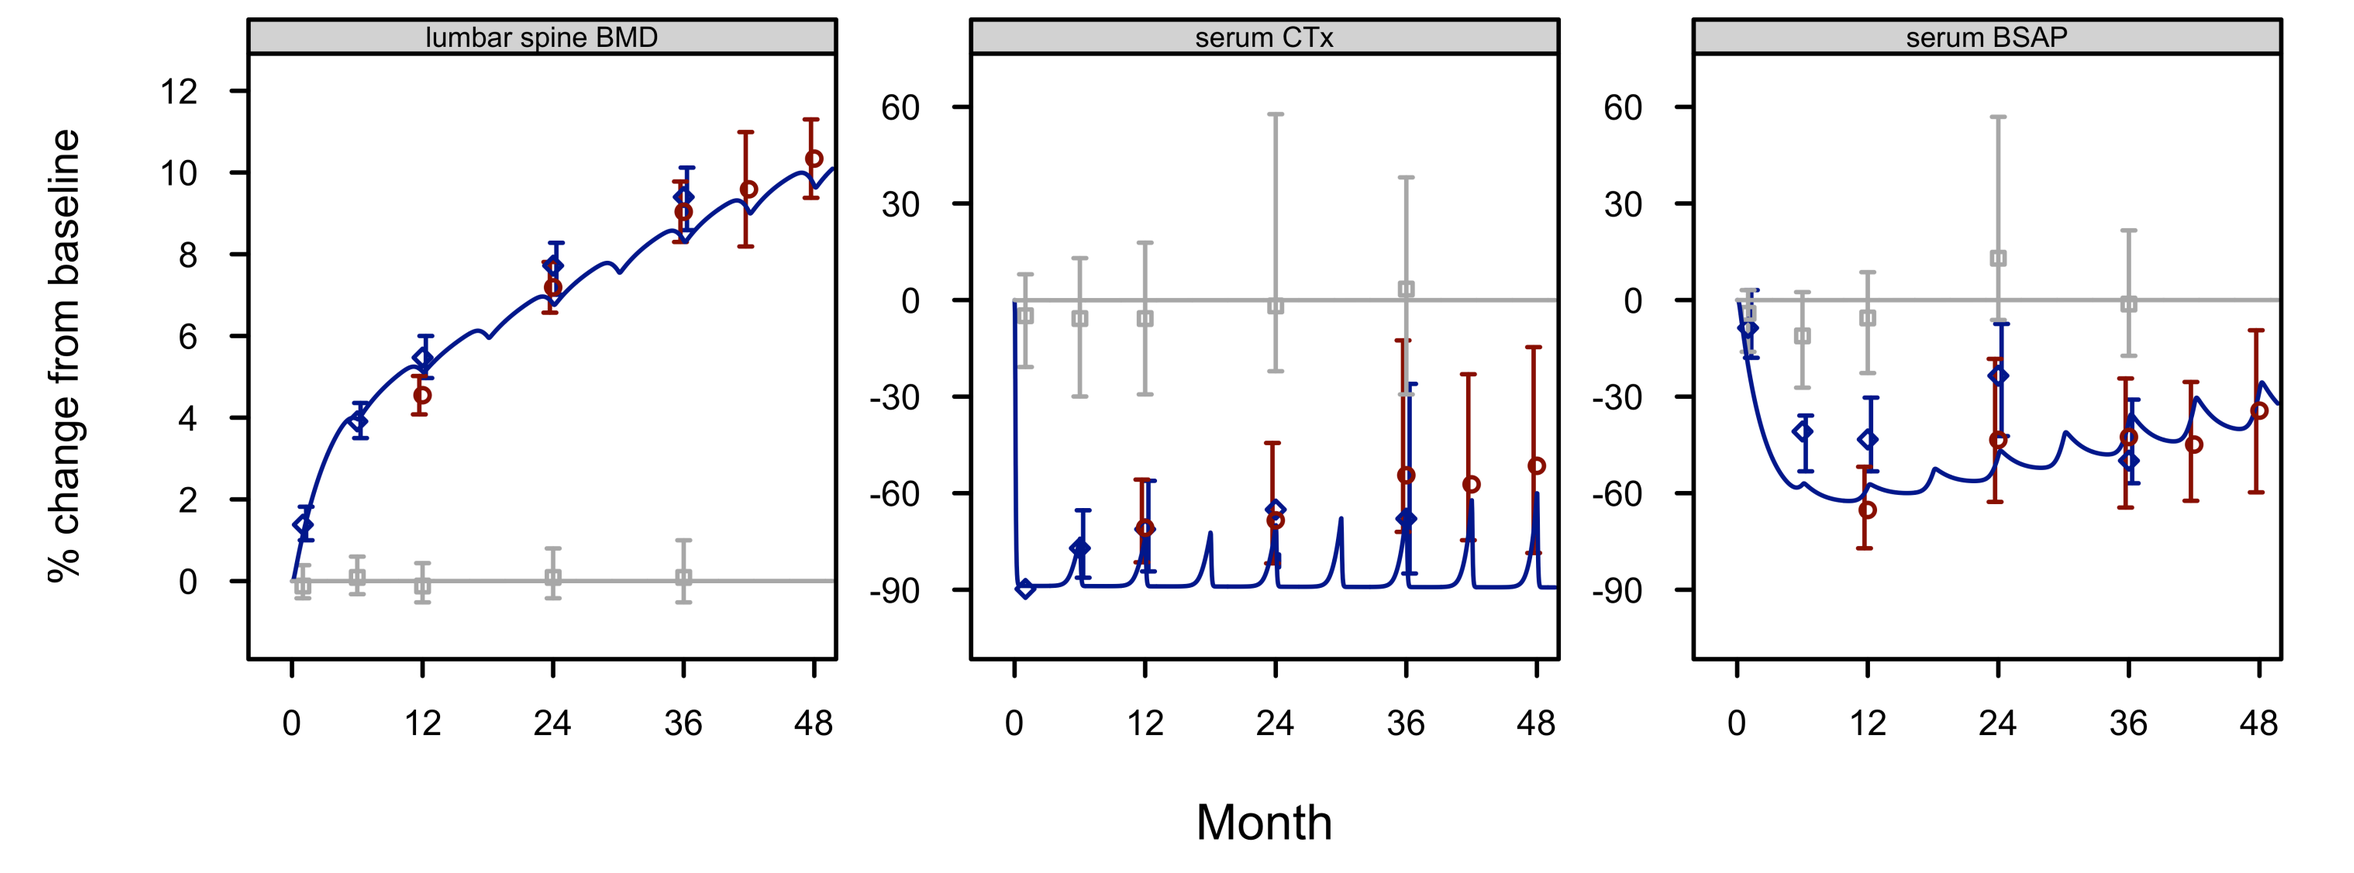
lumbar spine BMD
serum CTx
serum BSAP
% change from baseline
Month

Figure 6: *Observed (symbols) and simulated (lines) BMD, CTx, and BSAP during treatment with 60mg Q6M denosumab. Observed values from denosumab treatment groups: NCT00043186*[10, 15]*(red symbols, for model estimation); NCT00089791 (FREEDOM)*[13, 14] *(blue symbols, for denosumab model qualification) and placebo (grey symbols for reference).*

- MSPM predictions were in close agreement with observed data: Mean absolute percentage error = 9.1%; Mean percentage error = −7.9%
- MSPM predicted near complete decline in osteoclast function with slight increase in pre-dose (q 6 month) bone markers with continued administration

RESULTS – QUANTIFIED EFFICACY / SAFETY TARGET (EXAMPLE II)

ESSS (from logistic regression model); BMD (from MSPM)[3]

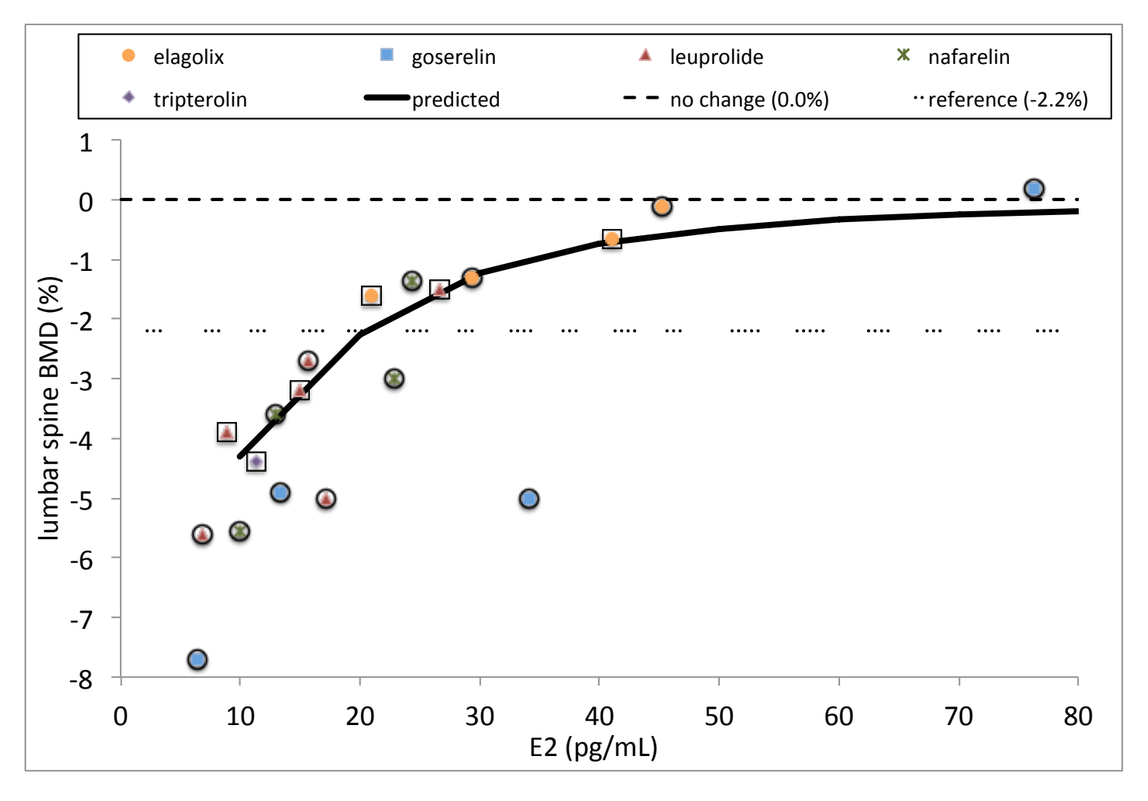
elagolix
goserelin
leuprolide
nafarelin
tripterolin
predicted
no change (0.0%)
reference (-2.2%)
lumbar spine BMD (%)
E2 (pg/mL)

Figure 7: *Observed (evaluation set) and Simulated BMD versus E2*

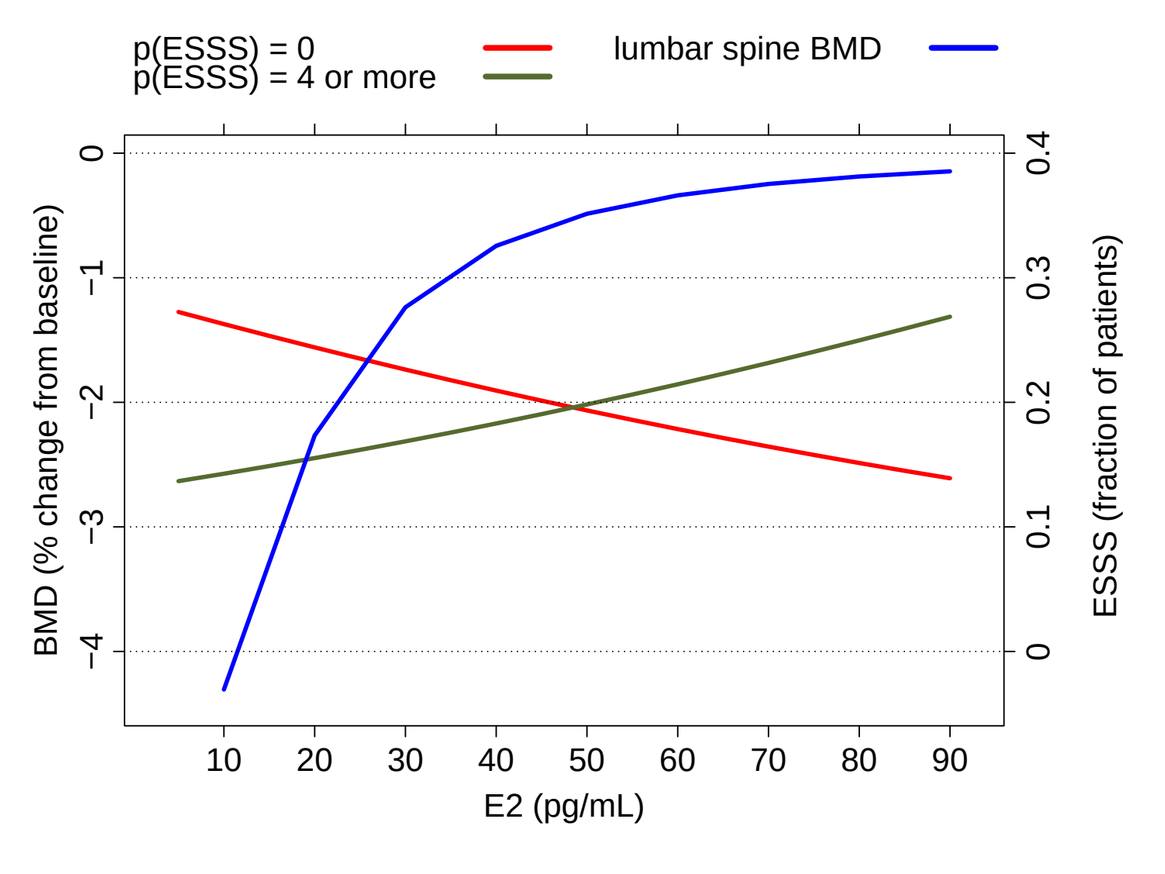
p(ESSS) = 0
p(ESSS) = 4 or more
lumbar spine BMD
BMD (% change from baseline)
ESSS (fraction of patients)
E2 (pg/mL)

Figure 8: *Comparison of Predicted ESSS and BMD versus E2*

- E2, measured as early as 1–2 months after treatment initiation, a reliable predictor of 6-month BMD change
- GnRH receptor modulation targeting E2 in the range of > 20 to 40 pg/mL expected to provide efficacious EM pain response while minimizing BMD effects (*Figure 8*)

RESULTS – LINKING BMD AND FRACTURE RISK (EXAMPLE III)

"Middle-out" MSPM Expansion to Predict Fracture Risk Clinical Outcome[8]

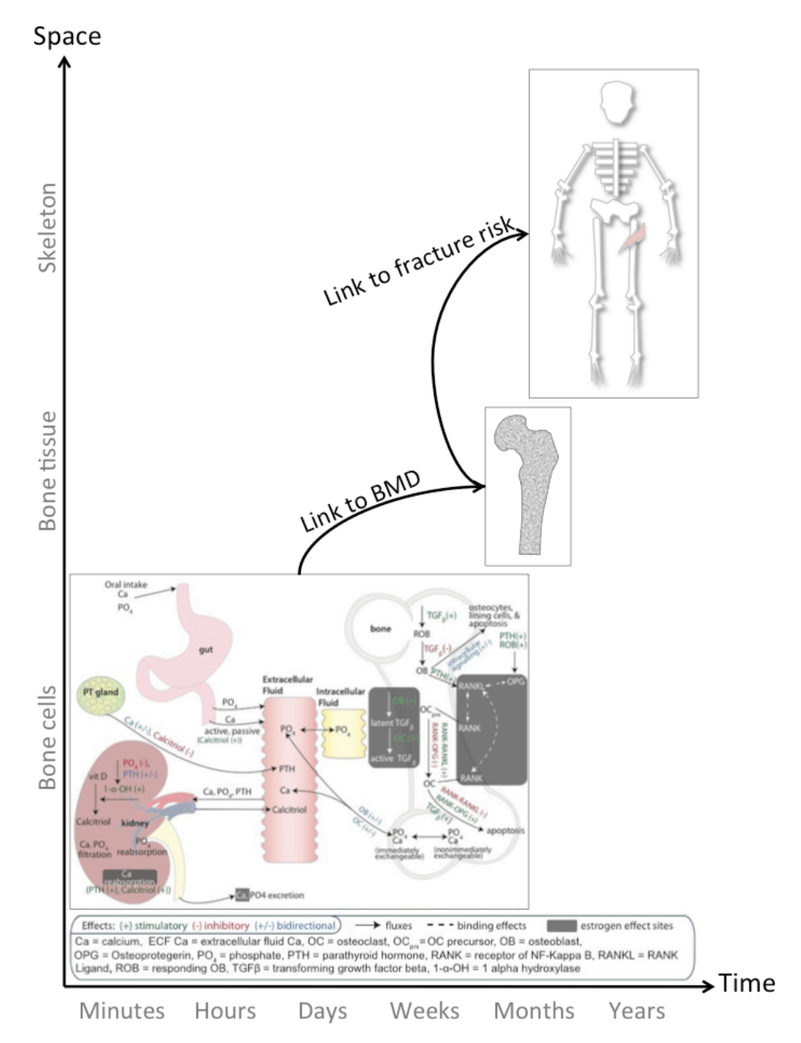
Space
Skeleton
Bone tissue
Bone cells
Link to fracture risk
Link to BMD
Minutes
Hours
Days
Weeks
Months
Years
Time

Figure 9: *MSPM extension to fracture risk*

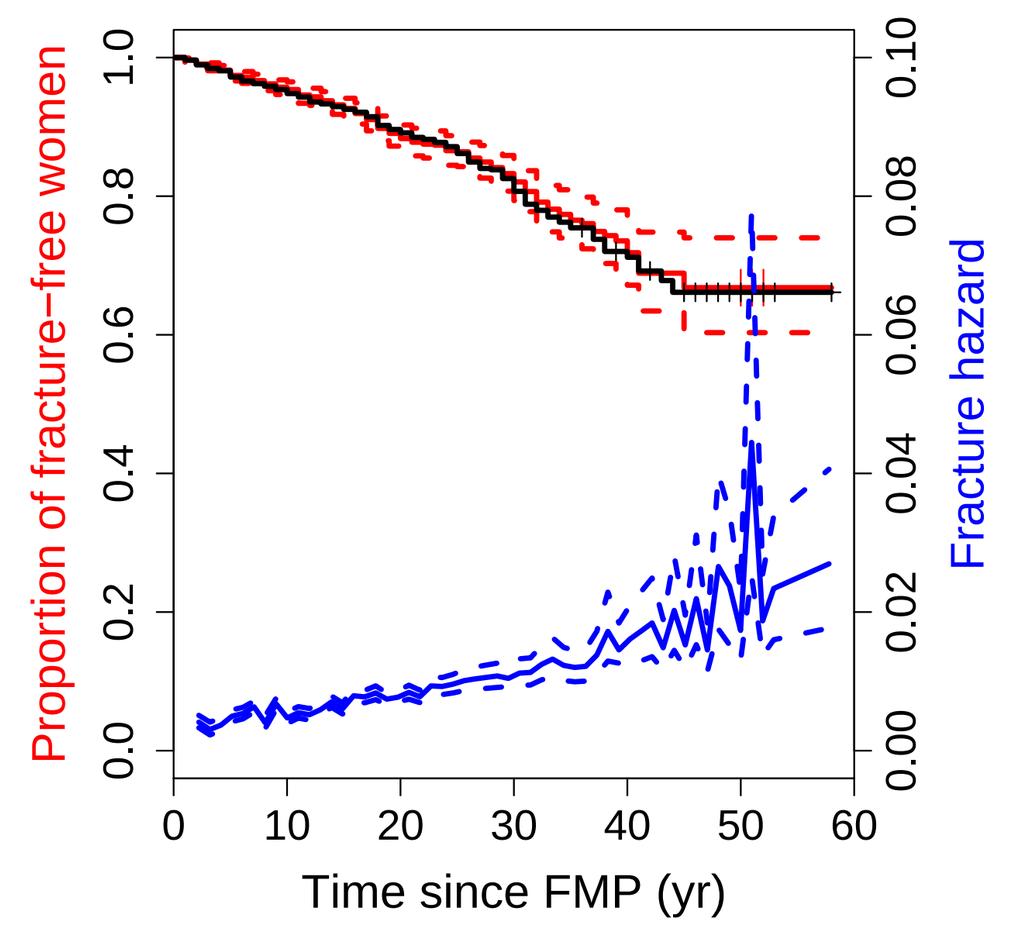
Proportion of fracture−free women
Fracture hazard
Time since FMP (yr)

Figure 10: *Simulated fracture hazard and survival*

- FMP_{age} and *time* were indirect covariates with dynamically predicted BMD
- Time-varying hazard reflects increase due to time-dependent BMD decline in final model
- Next steps include development of links with individual drug-relating effects, external evaluations, and consideration of site-specific fracture risks

SUMMARY – MSPM DEVELOPMENT TIMELINE

MSPM as an expandable R&D platform

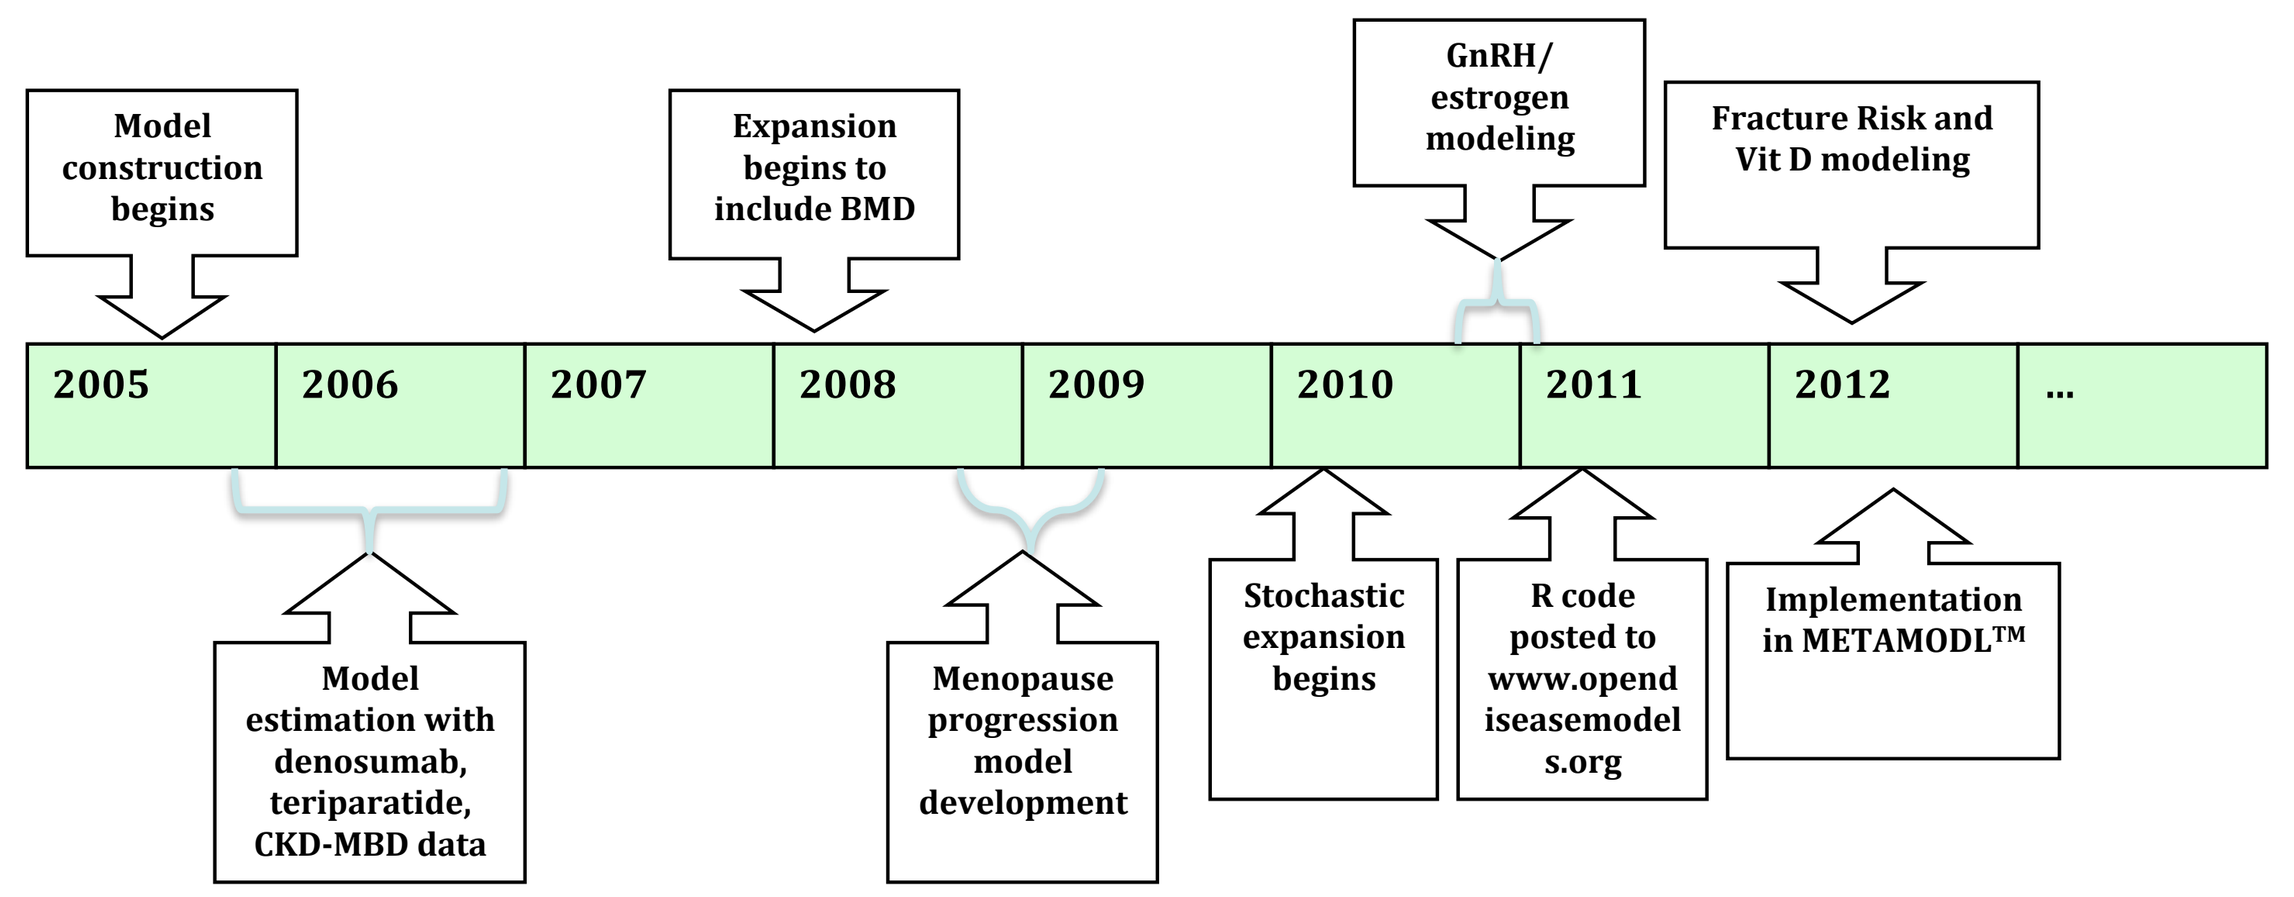
Model construction begins
Expansion begins to include BMD
GnRH/ estrogen modeling
Fracture Risk and Vit D modeling
2005
2006
2007
2008
2009
2010
2011
2012
...
Model estimation with denosumab, teriparatide, CKD-MBD data
Menopause progression model development
Stochastic expansion begins
R code posted to www.opendiseasemodels.org
Implementation in METAMODL™

CONCLUSIONS

- An MSPM can be constructed in an adaptable framework to address broad, but systems-related, research questions
- A well-founded MSPM offers efficient, timely extension and application
- MSPMs can serve as a repository of known mechanisms, hypotheses (theory), assumptions, and ongoing R&D goals
- MSPMs' ability to predict multiple therapeutic conditions provides further confidence in model-based predictions for decision-making in drug development and clinical practice

REFERENCES

[1] Peterson, M.C. and Riggs, M.M. A physiologically based mathematical model of integrated calcium homeostasis and bone remodeling. *Bone* **46** (2010):49–63.
[2] Riggs, M.M., Peterson, M.C. and Gastonguay, M.R. A systems biology model to describe long-term bone remodeling effects of estrogen in menopausal and postmenopausal women. *Clinical Pharmacology and Therapeutics* **85** (2009):S93.
[3] Riggs, M., Bennetts, M., van der Graaf, P. and Martin, S. Integrated pharmacometrics and systems pharmacology model-based analyses to guide GnRH receptor modulator development for management of endometriosis. *CPT Pharmacometrics Syst. Pharmacol.* **1** (2012).
[4] Riggs, M.M., Peterson, M.C. and Gastonguay, M.R. Multiscale physiology-based modeling of mineral bone disorder in patients with impaired kidney function. *J Clin Pharmacol* **52** (2012):45S–53S.
[5] Riggs, M.M. Multiscale systems models as a knowledge bridge between biology, physiology, and pharmacology. *AAPS Newsmagazine* (2011).
[6] Peterson, M.C. and M, R.M. Predicting nonlinear changes in bone mineral density over time using a multiscale systems pharmacology model. *CPT: pharmacomet. syst. pharmacol.* **1** (2012/11/14/online).
[7] Riggs, M.M., Baron, K.T., Plan, E.L. and Gastonguay, M.R. Qualification of a physiologically-based model for predicted bone marker and bone mineral density changes associated with denosumab treatment. In *Presented at American Society of Bone Mineral Research (ASBMR) Annual Meeting*, Abstract SU0363 (Minneapolis, MN, 2012).
[8] Plan, E.L., Baron, K.T., Gastonguay, M.R., French, J.L., Gillespie, W.R. and Riggs, M.M. Bayesian joint modeling of bone mineral density and repeated time-to-fracture event for multiscale bone systems model extension. In *PAGE 21st Meeting* (2012).
[9] R Development Core Team. *R: A Language and Environment for Statistical Computing*. R Foundation for Statistical Computing, Vienna, Austria (2008). ISBN: 3-900051-07-0. URL `http://www.R-project.org`
[10] McClung, M.R., Lewiecki, E.M., Cohen, S.B., Bolognese, M.A., Woodson, G.C., Moffett, A.H., Peacock, M., Miller, P.D., Lederman, S.N., Chesnut, C.H., Lain, D., Kivitz, A.J., Holloway, D.L., Zhang, C., Peterson, M.C., Bekker, P.J. and AMG 162 Bone Loss Study Group. Denosumab in postmenopausal women with low bone mineral density. *N Engl J Med* **354** (2006):821–31.
[11] Centers for Disease Control and Prevention (CDC), National Center for Health Statistics (NCHS). National health and nutrition examination survey data (nhanes).
[12] Cox, E.H., Veyrat-Follet, C., Beal, S.L., Fuseau, E., Kenkare, S. and Sheiner, L.B. A population pharmacokinetic-pharmacodynamic analysis of repeated measures time-to-event pharmacodynamic responses: the antiemetic effect of ondansetron. *Journal of Pharmacokinetics and Biopharmaceutics* **27** (1999):625–644.
[13] Cummings, S.R., San Martin, J., McClung, M.R., Siris, E.S., Eastell, R., Reid, I.R., Delmas, P., Zoog, H.B., Austin, M., Wang, A., Kutilek, S., Adami, S., Zanchetta, J., Libanati, C., Siddhanti, S., Christiansen, C. and FREEDOM Trial. Denosumab for prevention of fractures in postmenopausal women with osteoporosis. *N Engl J Med* **361** (2009):756–65.
[14] Eastell, R., Christiansen, C., Grauer, A., Kutilek, S., Libanati, C., McClung, M.R., Reid, I.R., Resch, H., Siris, E., Uebelhart, D., Wang, A., Weryha, G. and Cummings, S.R. Effects of denosumab on bone turnover markers in postmenopausal osteoporosis. *J Bone Miner Res* **26** (2011):530–7.
[15] Miller, P.D., Bolognese, M.A., Lewiecki, E.M., McClung, M.R., Ding, B., Austin, M., Liu, Y. and San Martin, J. Effect of denosumab on bone density and turnover in postmenopausal women with low bone mass after long-term continued, discontinued, and restarting of therapy: a randomized blinded phase 2 clinical trial. *Bone* **43** (2008):222–229.

Abstract #QSP-11: Presented at American Society of Clinical Pharmacology and Therapeutics (ASCPT) Annual Meeting and Quantitative Systems Pharmacology: An Integrating Framework for Translational Medicine Pre-Conference; Indianapolis, IN, 5-March-2013

Copies available at: www.metrumrg.com/publications

© Metrum Research Group LLC 2013