Qualification of a Physiologically-Based Model for Predicted Bone Marker and Bone Mineral Density Changes Associated with Denosumab Treatment

Matthew M. Riggs, Kyle T. Baron, Elodie L. Plan, Marc R. Gastonguay

Metrum Research Group LLC; 2 Tunxis Road Suite 112; Tariffville, CT USA

ABSTRACT

The goal of this investigation was to qualify the predictive performance of an existing physiologically-based, multiscale systems pharmacology model (MSPM) using data external to those used to develop it.

The MSPM was initially developed to describe longitudinal bone turnover marker (BTM) changes affected by both teriparatide and denosumab (dmab) treatment[1] and includes other relevant factors (e.g., calcium, PTH, calcitriol) and cellular and organ-level regulations, e.g., PTH control of urinary calcium excretion, RANK-RANKL-OPG system in bone, and differentiation and apoptosis controls for osteoclasts and osteoblasts. In the model, RANKL depletion affects an immediate decline in osteoclasts due to increased apoptosis and a later, less pronounced, decline in osteoblast function regulated by PTH and TGF-β changes. Development of this model included earlier dmab trial data (NCT00043186). A further extension of this model, using the earlier dmab data along with clinical manifestations associated with estrogen loss and during CKD-MBD ,[2] has linked bone markers with BMD change.

The external data used for the current evaluation were digitized from FREEDOM trial (NCT00089791) reports: serum C-terminal telopeptide (CTx), bone specific alkaline phosphatase (BSAP)[3] and lumbar spine (LS) BMD[4] . BTM and BMD model predictions over time were obtained for placebo (no intervention) and dmab 60 mg Q 6 months (for up to 4 years) and compared to the corresponding observed data.

Model predicted changes in LS BMD following dmab 60 mg Q6 months were: 1.1, 4.0, 5.2, 6.1, 6.9, 8.5, 9.2, and 9.8% at 1, 6, 12, 18, 24, 36, 42 and 48 months, respectively, and were in close agreement with observed data: mean absolute percentage error=9.1%; mean percentage error=-7.9%. The model also predicted the nearly complete decline in osteoclast function with a slight increase in pre-dose (6 months after previous dose) CTx and BSAP with continued administration.

The use of external data to qualify the performance of the existing MSPM for prediction of BTM and BMD changes associated with dmab treatment indicated that the model predictions can be generalized across data sets. These results provide further confidence in model-based predictions of physiologic changes due to modulation of the RANK-L system and related decision-making in drug development and clinical practice.

INTRODUCTION – MODEL BACKGROUND

Multiscale Systems Biology / Pharmacology Models (Figure 1)

- Biologic systems expressed as mathematical expressions
- Quantify timecourses, magnitudes of changes (e.g., natural decays, interactions)
- Serve as *in silico* probes of biologic perturbation (e.g., disease, genetic variation)
- Multiscale systems pharmacology model (**MSPM**): include pharmacologic effects

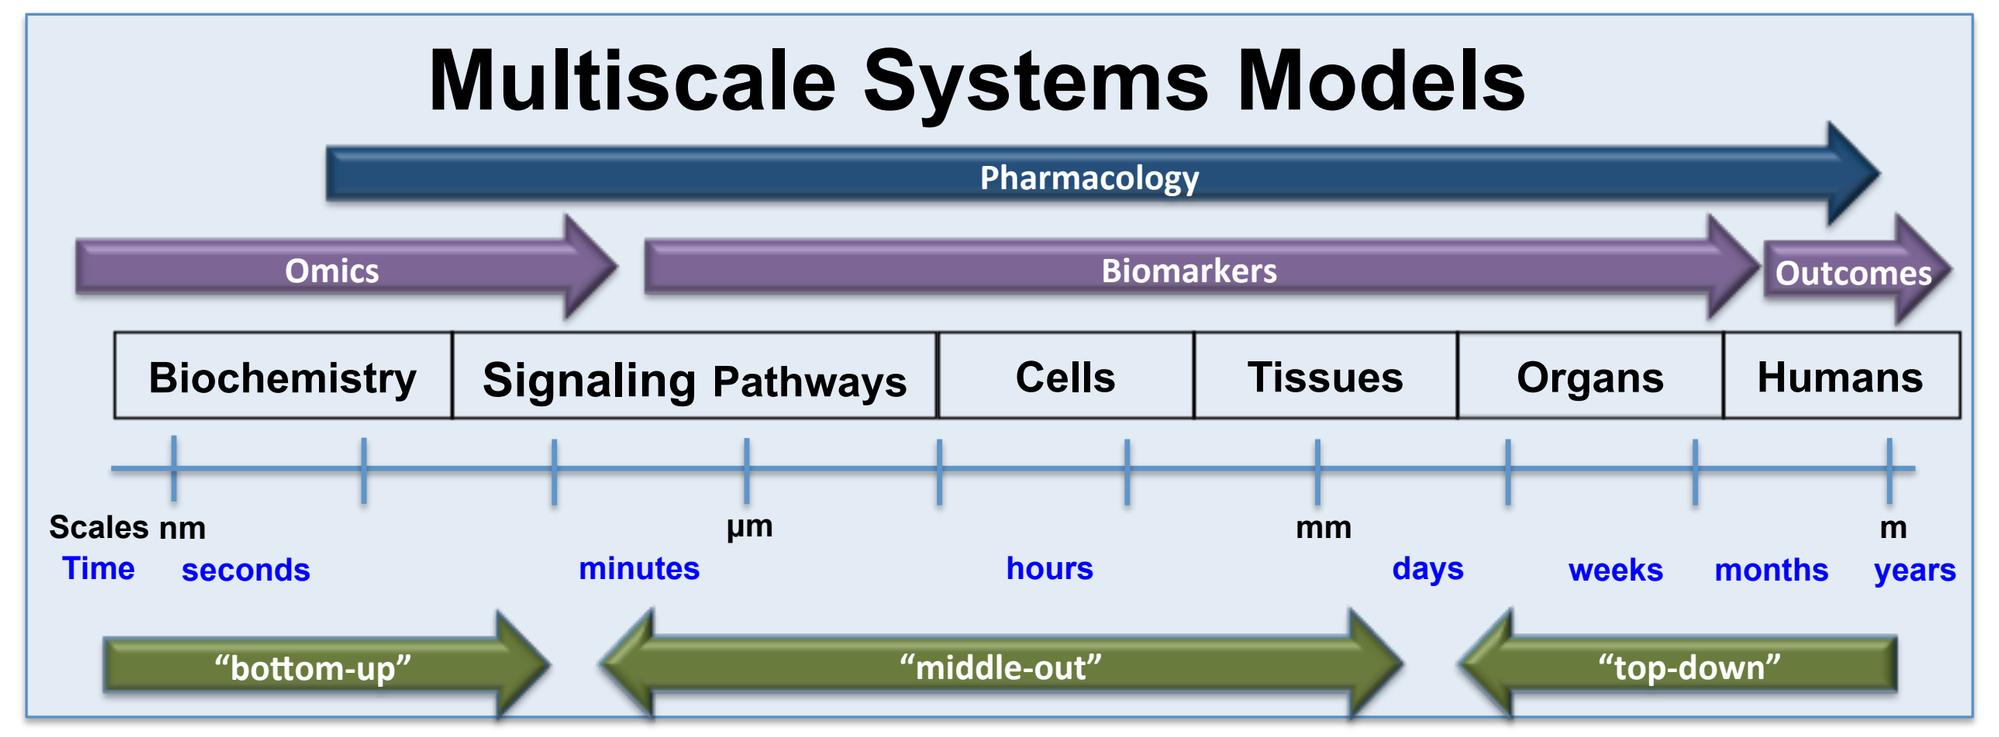
Multiscale Systems Models
Pharmacology
Omics
Biomarkers
Outcomes
Biochemistry
Signaling Pathways
Cells
Tissues
Organs
Humans
Scales nm
μm
mm
m
Time
seconds
minutes
hours
days
weeks
months
years
"bottom-up"
"middle-out"
"top-down"

Figure 1: *Defining multiscale systems models and terminology; reproduced from Riggs 2011[5]*

MSPM of Bone Mineral Homeostasis and Remodeling (Figures 2, 3)

- Mathematical (differential equations) construct from experimental and clinical data
- Scales: Cell signaling → organ functions → bone turnover markers (BTMs) → BMD
- Applications:
 - Denosumab: PTH, serum calcium, BTMs,[1] and lumbar spine BMD[6]
 - Teriparatide: PTH, serum calcium, and BTMs[1]
 - Disease/Aging [CKD-MBD,[2] menopause and endometriosis[7]]: BTMs, BMD and fracture risk[8]
- Software: R (www.R-project.org/)[9]
- Model code available through:
 - Original publication[1] (open-source): www.opendiseasemodels.org
 - Ongoing development (subscription-based): METAMODL™ (www.metamodl.com)

Denosumab: inhibit one pathway (RANK-RANKL) ⇒ model related effects: osteoclast and osteoblast activity (BTMs), BMD, and peripheral effects, e.g., serum calcium and PTH.

OBJECTIVE: MSPM Qualification

- **Can the MSPM predict clinical observations that were not included in the original model development? In this case, endpoint data collected from a separate clinical study.**

INTRODUCTION – MULTISCALE SYSTEMS PHARMACOLOGY MODEL OVERVIEW

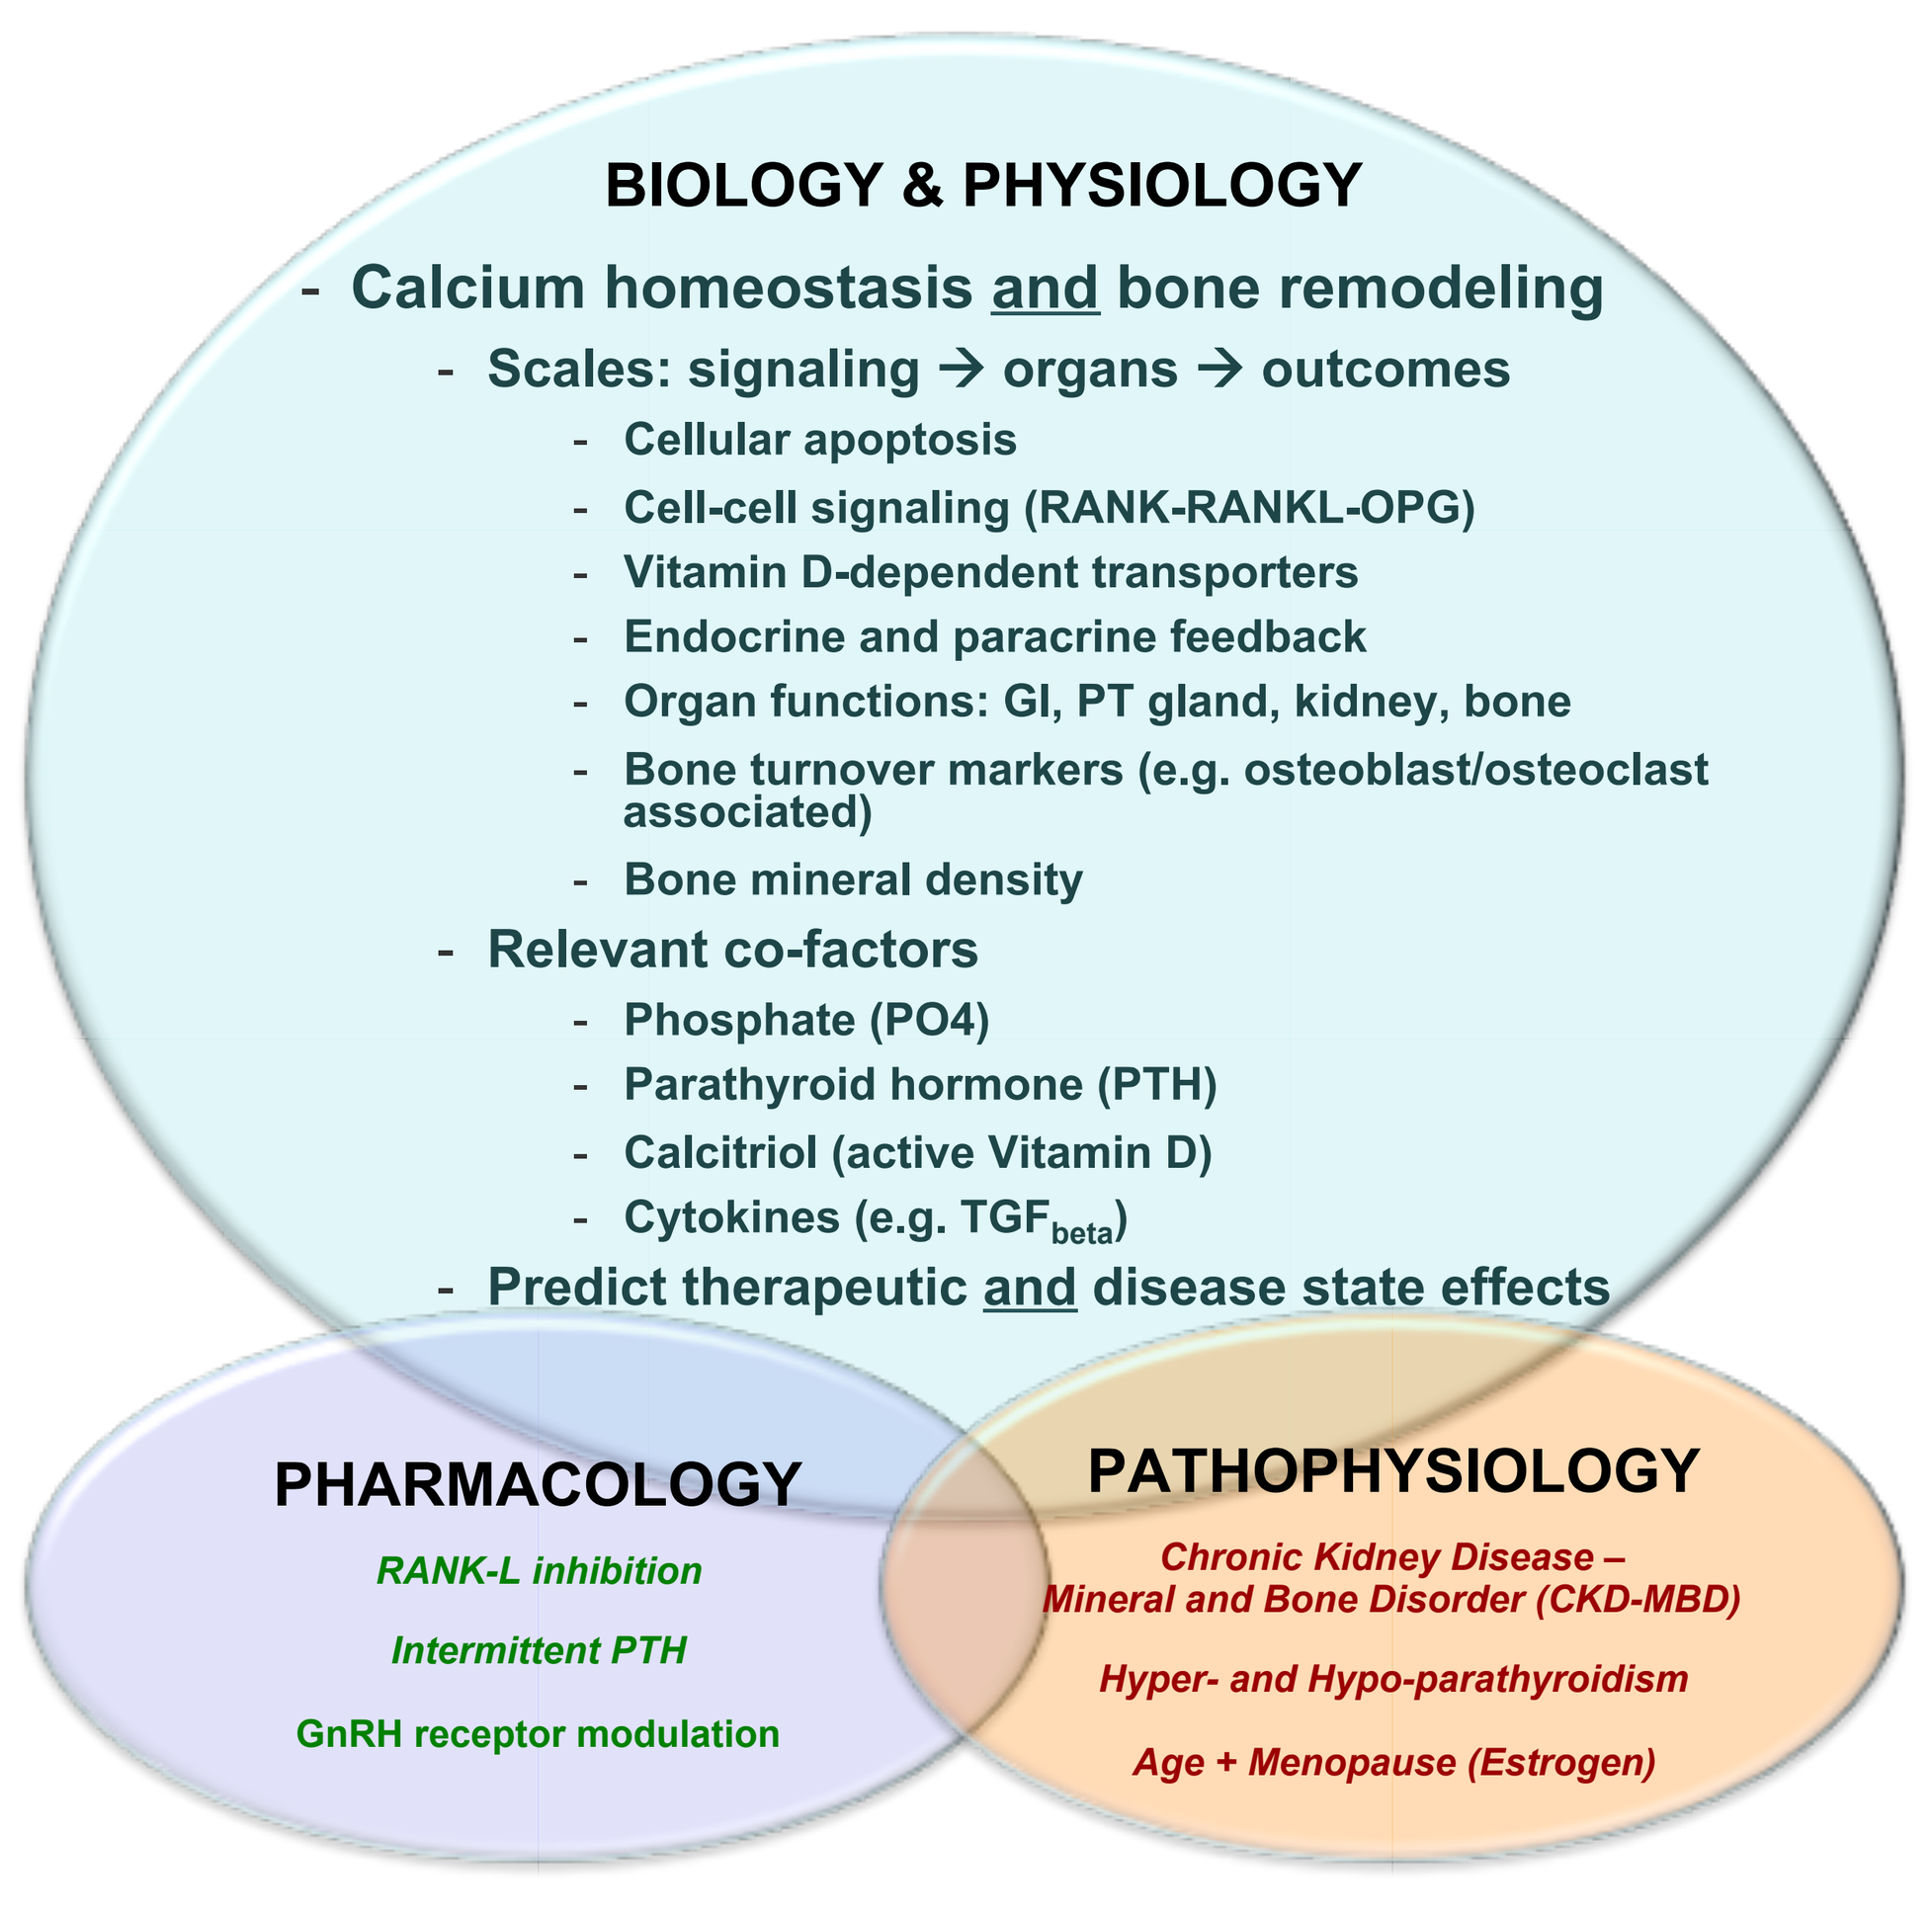
BIOLOGY & PHYSIOLOGY
- Calcium homeostasis and bone remodeling
- Scales: signaling → organs → outcomes
- Cellular apoptosis
- Cell-cell signaling (RANK-RANKL-OPG)
- Vitamin D-dependent transporters
- Endocrine and paracrine feedback
- Organ functions: GI, PT gland, kidney, bone
- Bone turnover markers (e.g. osteoblast/osteoclast associated)
- Bone mineral density
- Relevant co-factors
- Phosphate (PO4)
- Parathyroid hormone (PTH)
- Calcitriol (active Vitamin D)
- Cytokines (e.g. TGFbeta)
- Predict therapeutic and disease state effects
PHARMACOLOGY
RANK-L inhibition
Intermittent PTH
GnRH receptor modulation
PATHOPHYSIOLOGY
Chronic Kidney Disease – Mineral and Bone Disorder (CKD-MBD)
Hyper- and Hypo-parathyroidism
Age + Menopause (Estrogen)

Figure 2: *MSPM conceptual framework and content overview.*

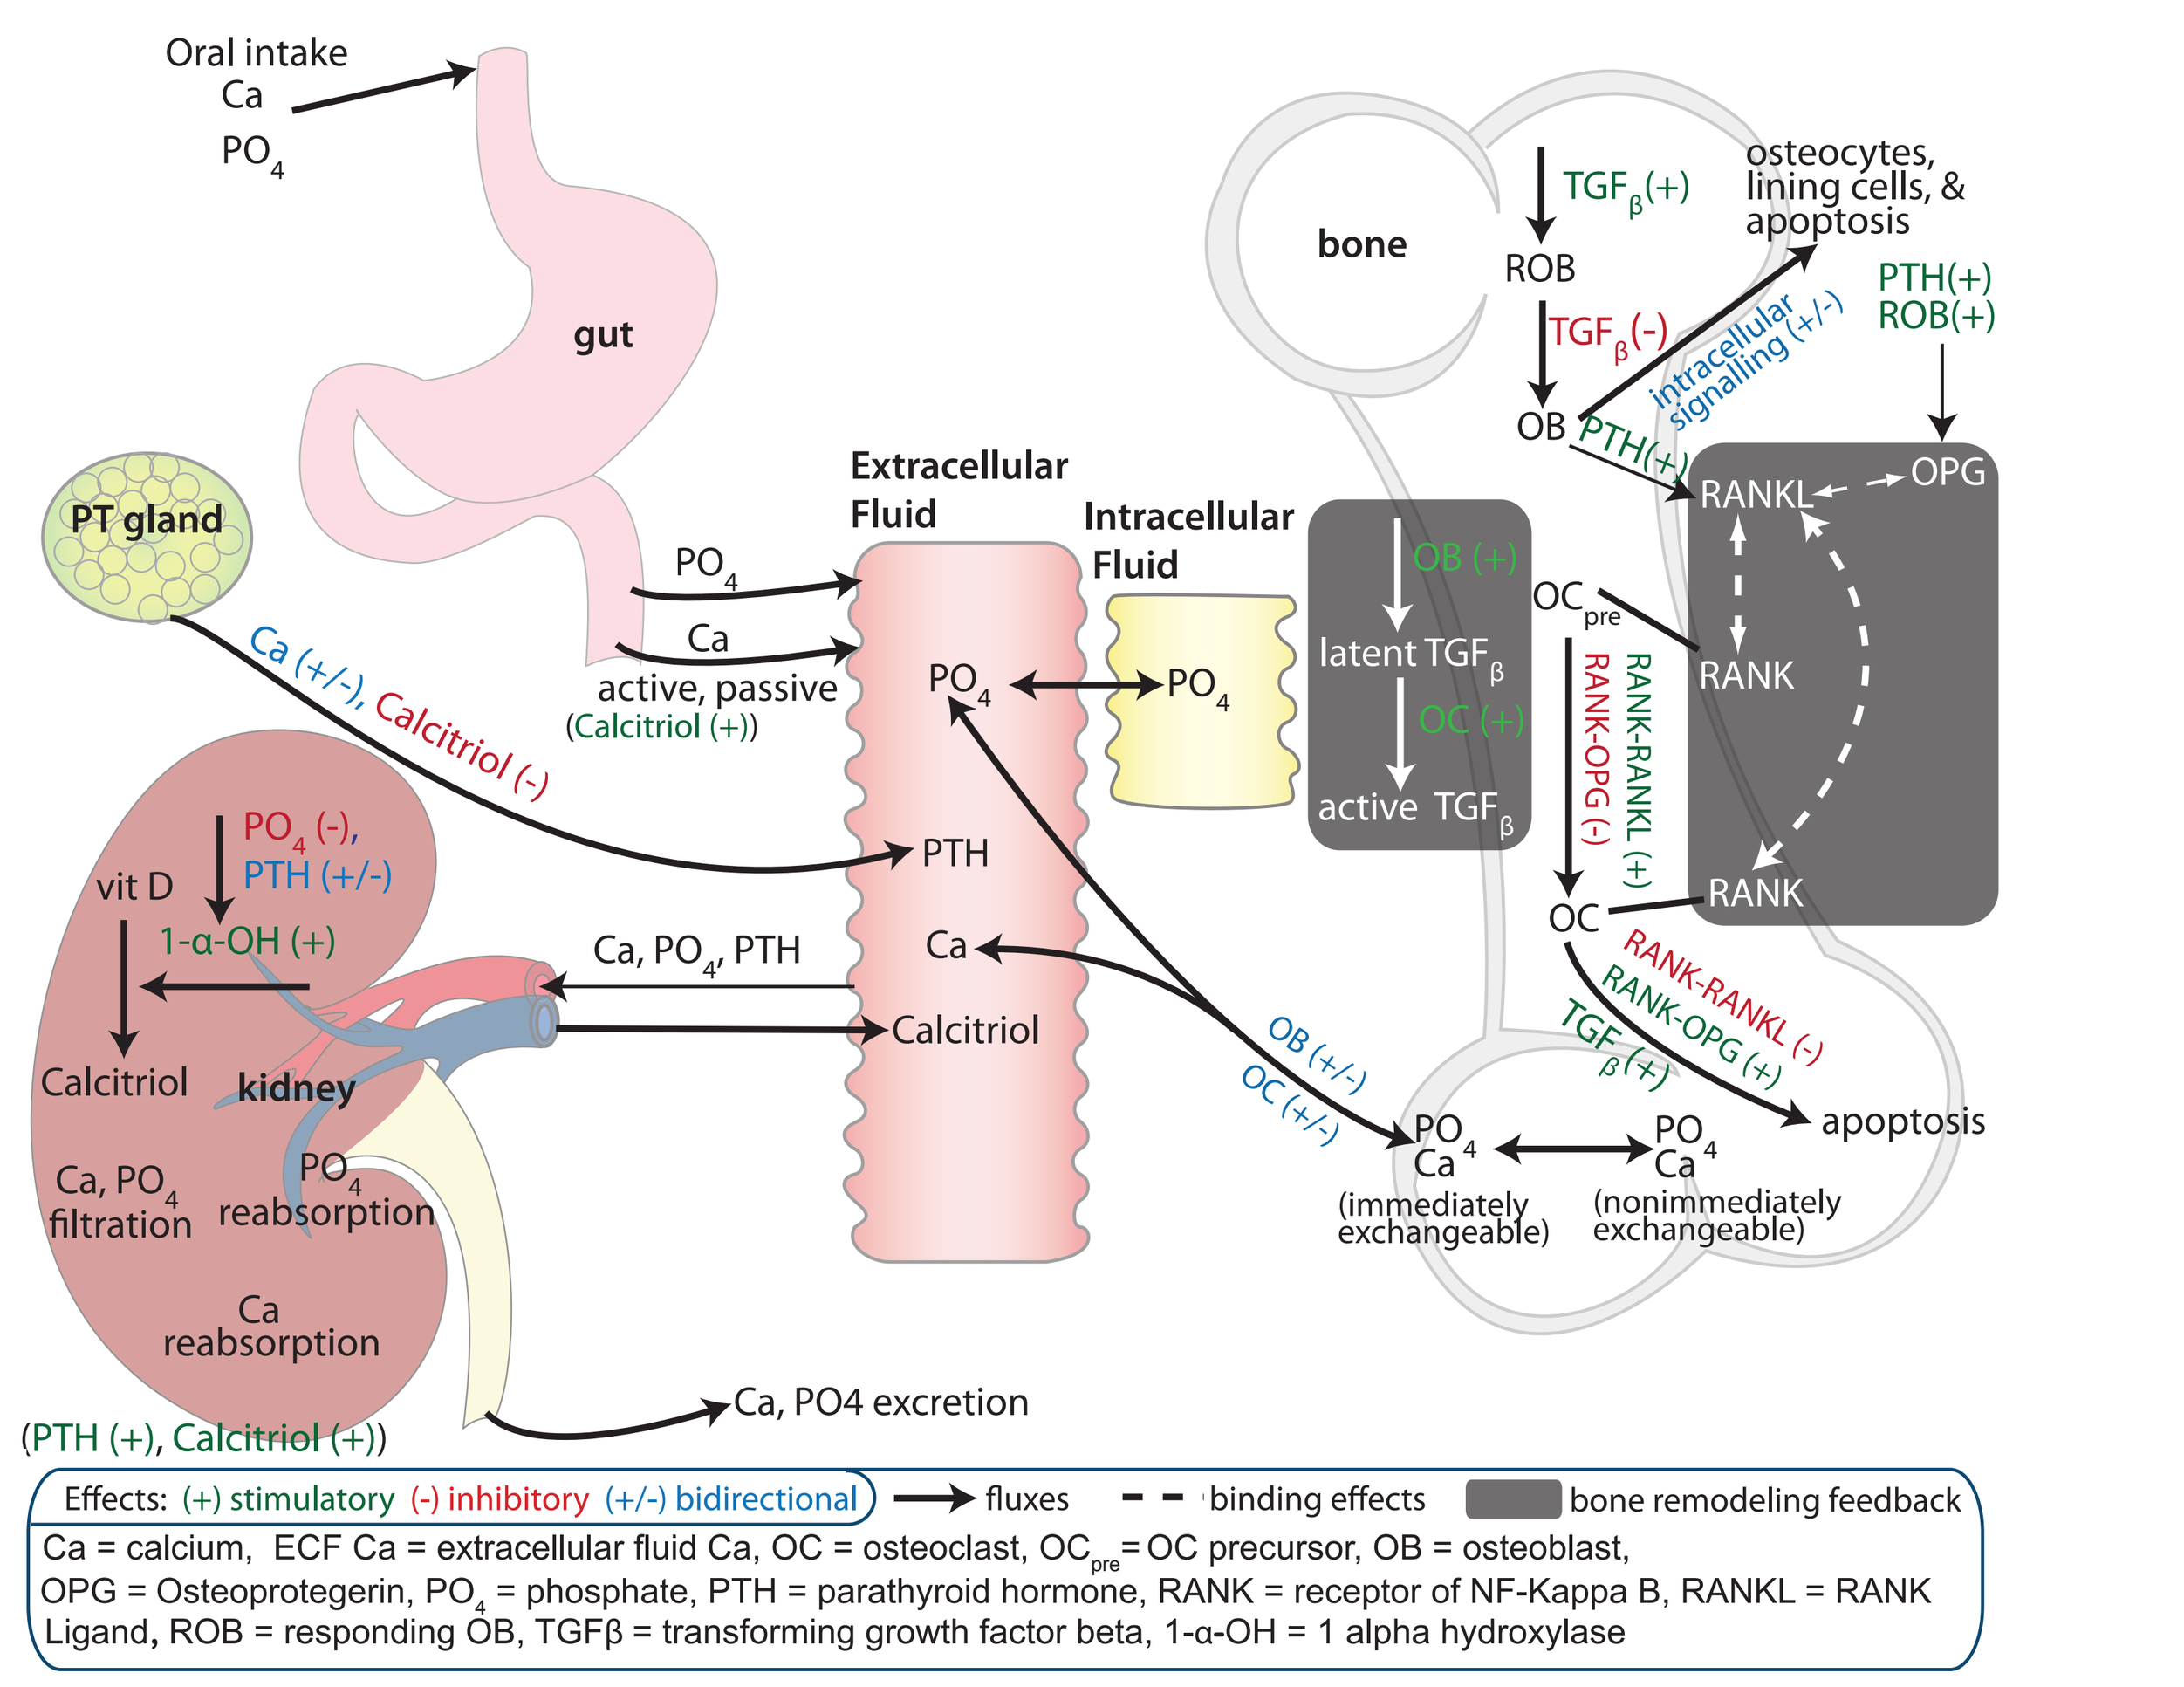
Oral intake
Ca
PO4
gut
PT gland
Ca (+/-), Calcitriol (-)
PO4
Ca
active, passive
(Calcitriol (+))
Extracellular Fluid
Intracellular Fluid
PO4
PTH
Ca
Calcitriol
vit D
PO4 (-), PTH (+/-)
1-α-OH (+)
Calcitriol
kidney
Ca, PO4, PTH
Ca, PO4 filtration
PO4 reabsorption
Ca reabsorption
Ca, PO4 excretion
(PTH (+), Calcitriol (+))
bone
TGFβ(+)
ROB
TGFβ(-)
OB
PTH(+)
intracellular signalling (+/-)
osteocytes, lining cells, & apoptosis
PTH(+) ROB(+)
OB (+)
latent TGFβ
OC (+)
active TGFβ
OCpre
RANKL
OPG
RANK
RANK-RANKL (+)
RANK-OPG (-)
RANK
OC
RANK-RANKL (-)
RANK-OPG (+)
TGFβ(+)
apoptosis
OB (+/-)
OC (+/-)
PO4 Ca (immediately exchangeable)
PO4 Ca (nonimmediately exchangeable)
Effects: (+) stimulatory (-) inhibitory (+/-) bidirectional → fluxes - - binding effects ■ bone remodeling feedback
Ca = calcium, ECF Ca = extracellular fluid Ca, OC = osteoclast, OCpre = OC precursor, OB = osteoblast,
OPG = Osteoprotegerin, PO4 = phosphate, PTH = parathyroid hormone, RANK = receptor of NF-Kappa B, RANKL = RANK
Ligand, ROB = responding OB, TGFβ = transforming growth factor beta, 1-α-OH = 1 alpha hydroxylase

Figure 3: *Schematic of physiologically-based, multiscale systems pharmacology model; modified from figure 1 of Peterson and Riggs, 2010.* [1]

METHODS – RANK-RANKL-OPG and DENOSUMAB

The Receptor Activator of Nuclear Factor-κ B (RANK)-RANK Ligand (RANKL)-Osteoprotegerin (OPG) system

⇑ OC differentiation and ⇑ OC activation: RANK–RANKL

⇓ OC differentiation and ⇑ OC apoptosis: RANKL–OPG, RANKL–denosumab

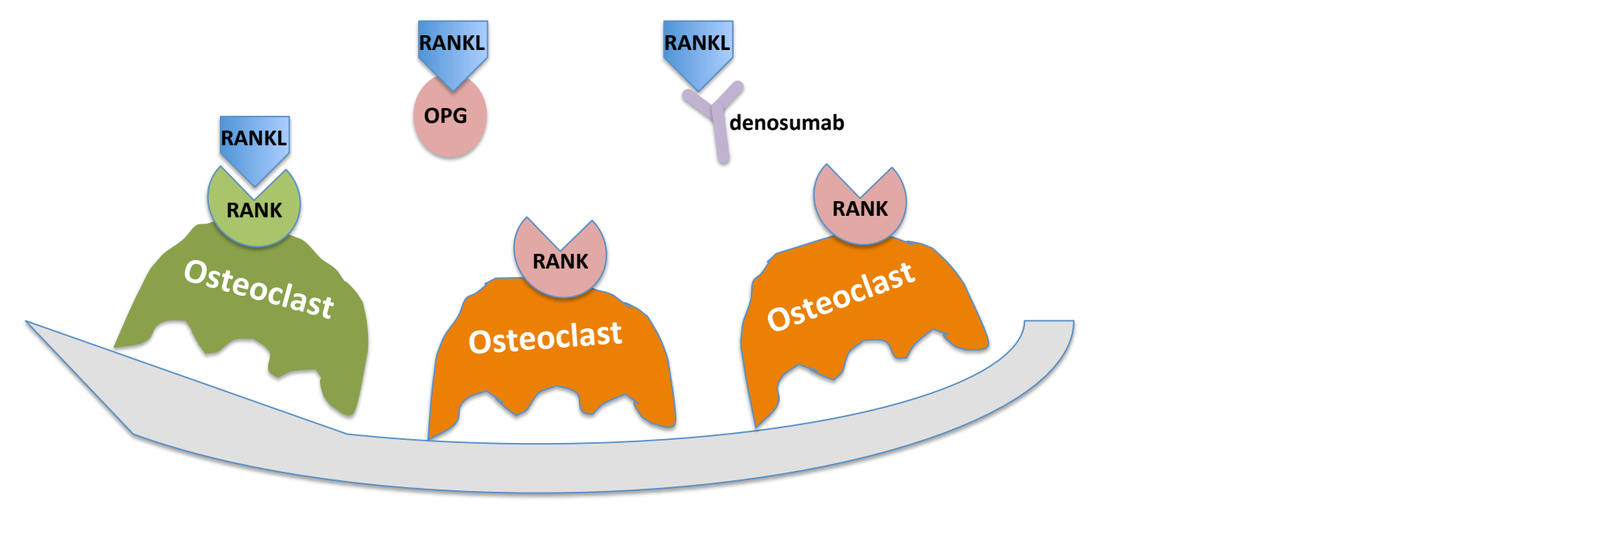
RANKL
RANK
Osteoclast
OPG
RANKL
RANK
Osteoclast
RANKL
denosumab
RANK
Osteoclast

Figure 4: *Depiction of RANK-RANKL-OPG and denosumab interaction.*

Denosumab[10]
- Fully human monoclonal antibody
- Binds to RANKL with high affinity and specificity
- Blocks interaction of RANKL with RANK
- Mimics endogenous effects of OPG

Denosumab–RANKL binding

- ↓ available RANKL
- ↓ RANK–RANKL interaction
- ↓ Osteoclast activity (serum C-telopeptide, CTx)
- ↓ Activation of TGFβ
- ↓ Osteoblast activity (bone-specific alkaline phosphatase, BSAP)
- ↑ bone mineral density (BMD), expressed mathematically[6] as:

$$\frac{d(LS\ BMD)}{dt} = \mathrm{k}_{\mathrm{in},BMD} \cdot \left(\frac{\mathrm{BSAP}}{\mathrm{BSAP}_{\mathrm{baseline}}}\right)^{\gamma_{OB}} - \mathrm{k}_{out,BMD} \cdot \left(\frac{\mathrm{CT_x}}{\mathrm{CT}_{\mathrm{x_{baseline}}}}\right)^{\gamma_{OC}} \cdot LS\ BMD$$

$$\gamma_{OB} = 0.0793 \quad \gamma_{OC} = 0.0679 \quad k_{\mathrm{out,BMD}} = 0.000145\ \mathrm{h}^{-1}$$

Denosumab pharmacokinetic model

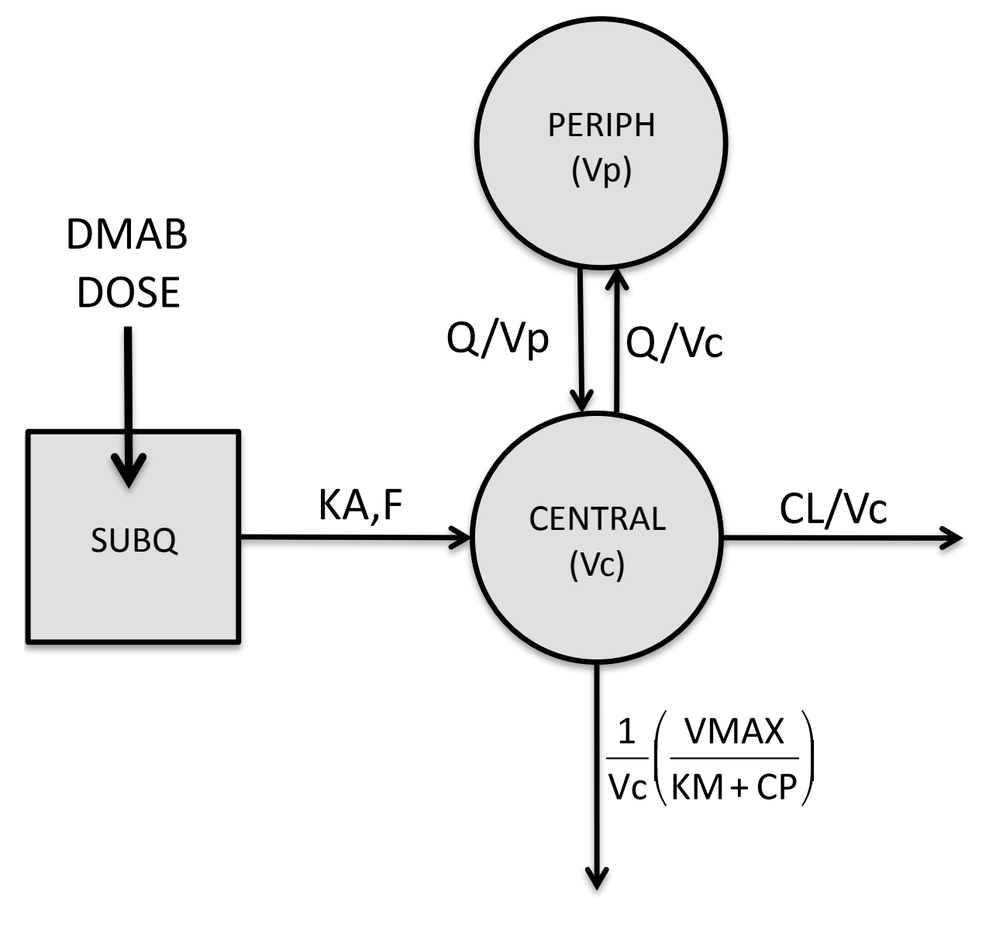
PERIPH (Vp)
DMAB DOSE
Q/Vp
Q/Vc
SUBQ
KA,F
CENTRAL (Vc)
CL/Vc
1/Vc (VMAX/(KM+CP))

Figure 5: *Denosumab pharmacokinetic model.*

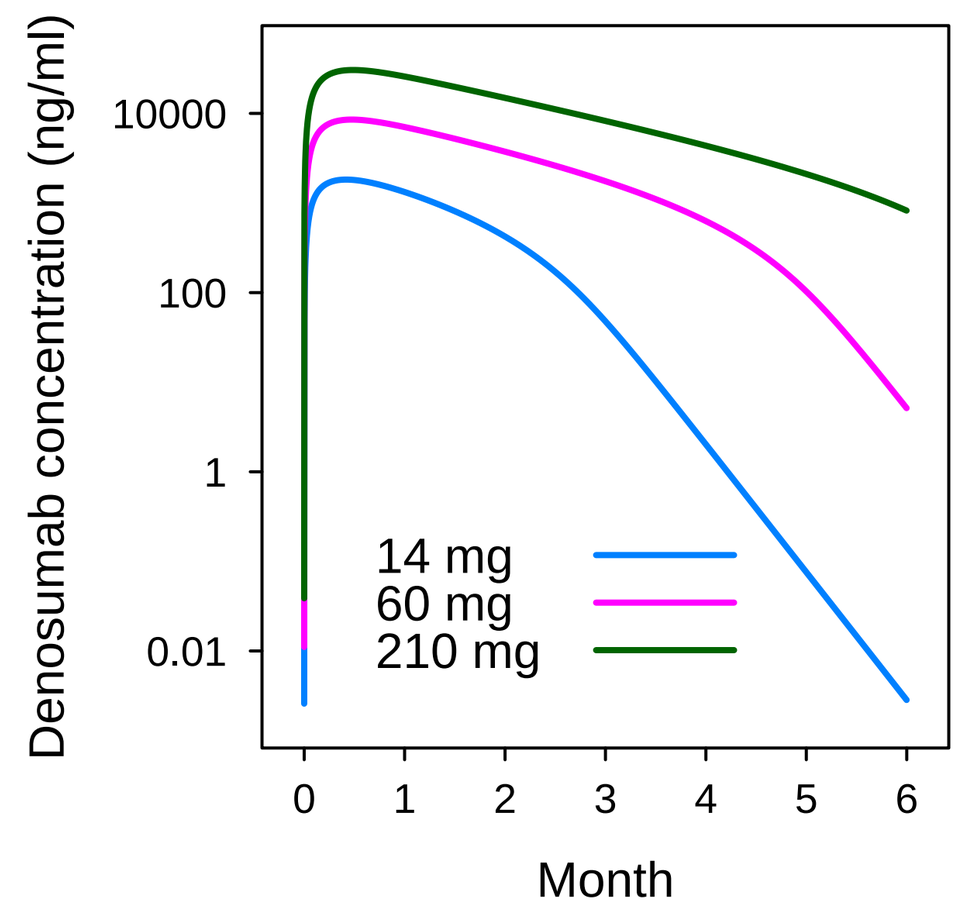
Denosumab concentration (ng/ml)
10000
100
1
0.01
14 mg
60 mg
210 mg
0 1 2 3 4 5 6
Month

Figure 6: *Simulated single-dose denosumab concentration versus time.*

CL	Clearance - linear process	2.75	ml hr^{-1}
Vc	Central volume of distribution	2340	ml
Vp	Peripheral volume of distribution	1324	ml
Q	Intercompartmental clearance	18.67	ml hr^{-1}
Ka	Absorption rate constant	0.00592	hr^{-1}
F	Bioavailability	0.729	–
Vmax	Maximum rate - nonlinear process	3110	ng hr^{-1}
Km	Michaelis constant	180	ng ml^{-1}

Table 1: *Denosumab pharmacokinetic parameters, from Peterson et al.[11]*

METHODS – DENOSUMAB CLINICAL DATA: From Two Randomized, Double-Blind, Placebo-controlled, Multi-dose Studies

NCT00043186: Postmenopausal Women With Low BMD[10, 12]

- **MSPM development and estimation dataset**
- 6 – 210 mg denosumab (as Q3M and Q6M regimens): up to 48 months
- Endpoints included: PTH, serum calcium, BTMs, and LS BMD

NCT00089791 (FREEDOM): Postmenopausal Osteoporosis[3, 4]

- **MSPM qualification dataset**
- 60 mg Q6M administered for up to 48 months
- Endpoints included: BTMs (CTx and BSAP)[3] & LS BMD[4]

METHODS – DENOSUMAB DOSING REGIMEN

For figures 7 through 9 : Dose and dosing interval for 0 to 24 months are given in the panel title. At 24 months, regimen was updated as follows: 60 mg Q6M had no change to the regimen; 14mg Q6M switched to 60mg Q6M for 24 months; 30 mg Q3M switch to placebo for 12 months, then 60mg Q6M for 12 months; 210 mg Q6M switched to placebo for 24 months.[12]

METHODS – MSPM DEVELOPMENT: BSAP, CTx, BMD

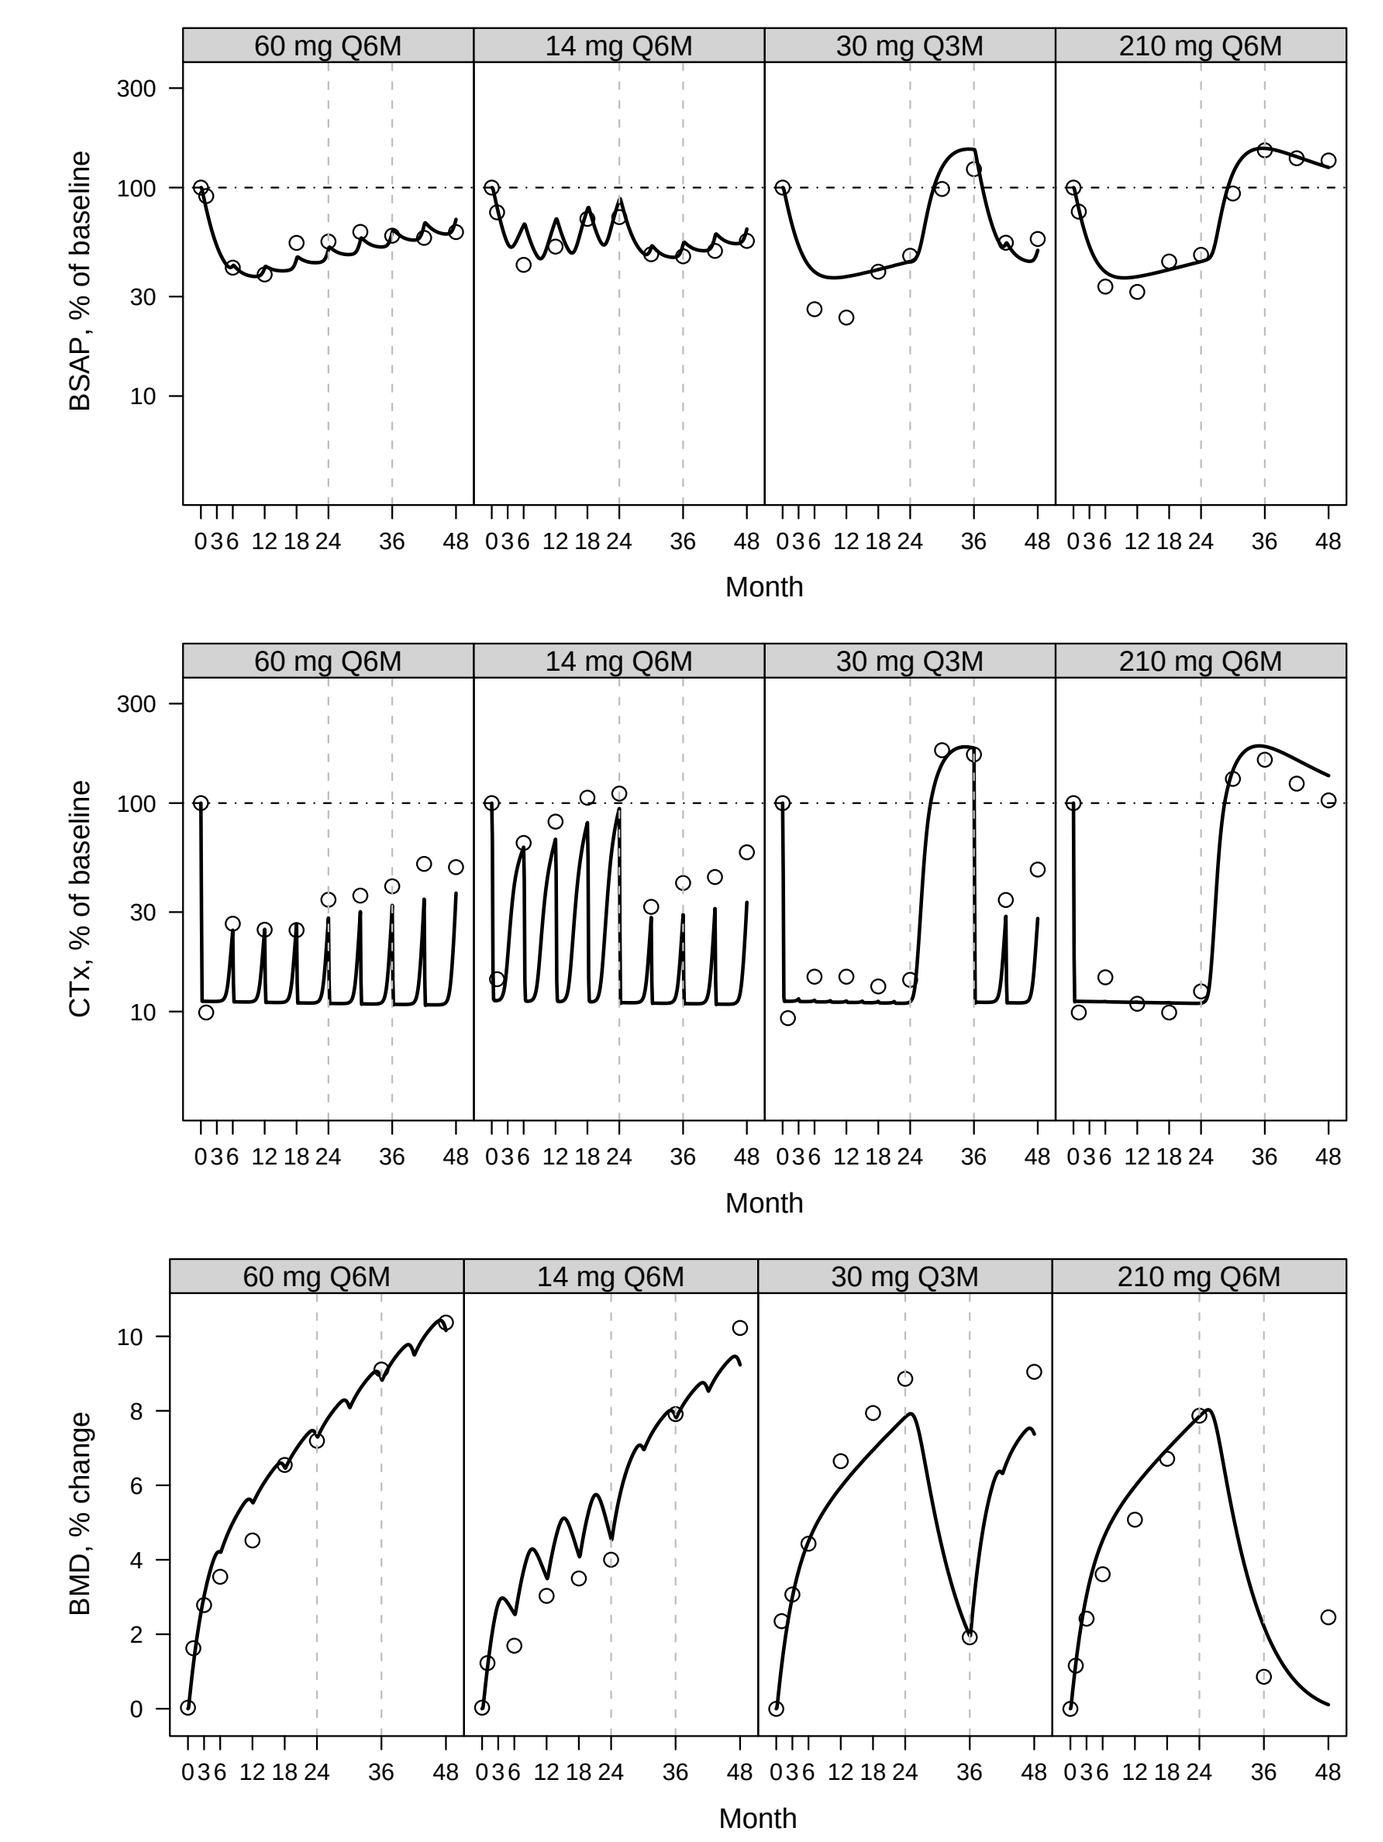
60 mg Q6M
14 mg Q6M
30 mg Q3M
210 mg Q6M
BSAP, % of baseline
CTx, % of baseline
BMD, % change
Month

Figure 7: *Observed (points)[10, 12] and MSPM predicted (lines) BSAP (top), CTx (middle), and lumbar spine BMD (bottom) following denosumab multiple dosing. Figures taken from Peterson and Riggs[6]*

RESULTS – MSPM EXPLORATION OF RANK-L & TGF-β EFFECTS

MSPM provides exploration of changes that are otherwise unmeasured

- Rebound in RANKL and TGF-β help explain CTx and BSAP drift with repeated dosing

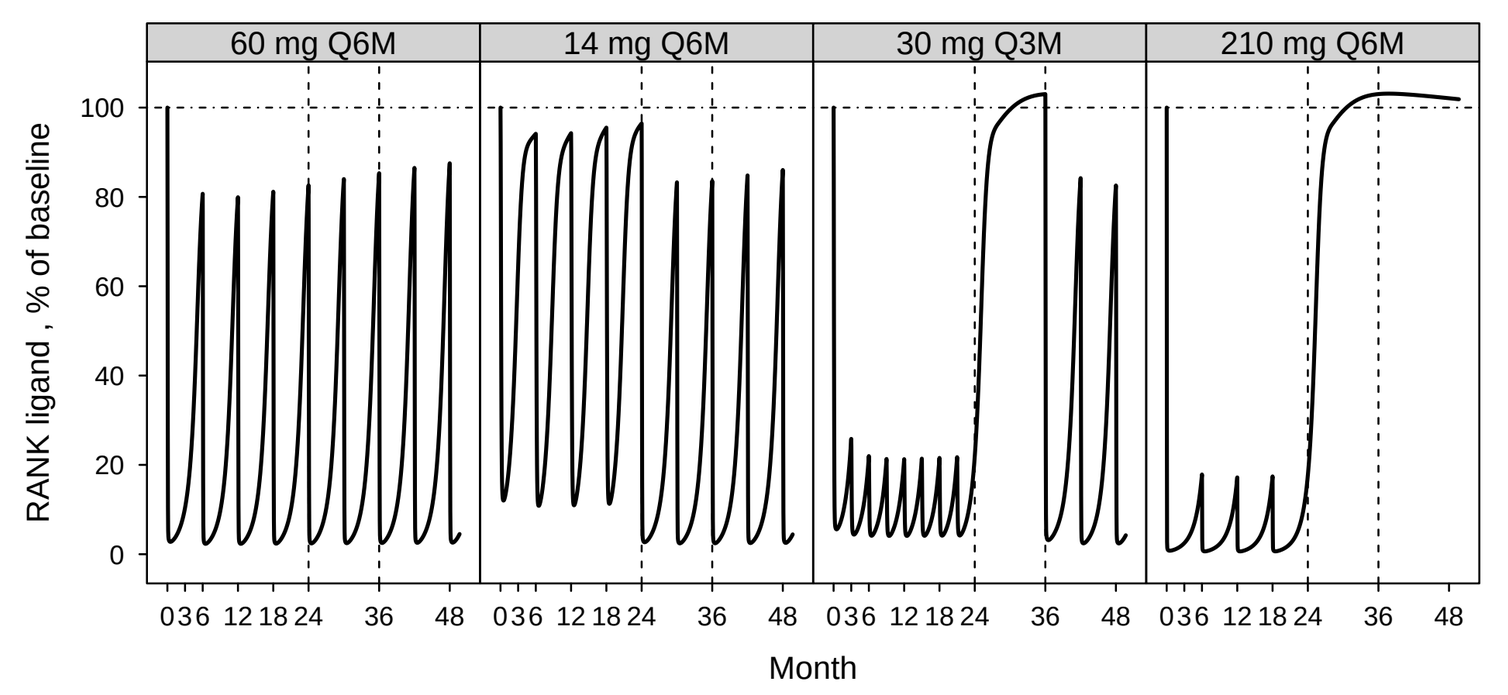
60 mg Q6M
14 mg Q6M
30 mg Q3M
210 mg Q6M
RANK ligand , % of baseline
Month

Figure 8: *MSPM simulated changes in RANK ligand after multiple denosumab doses.*

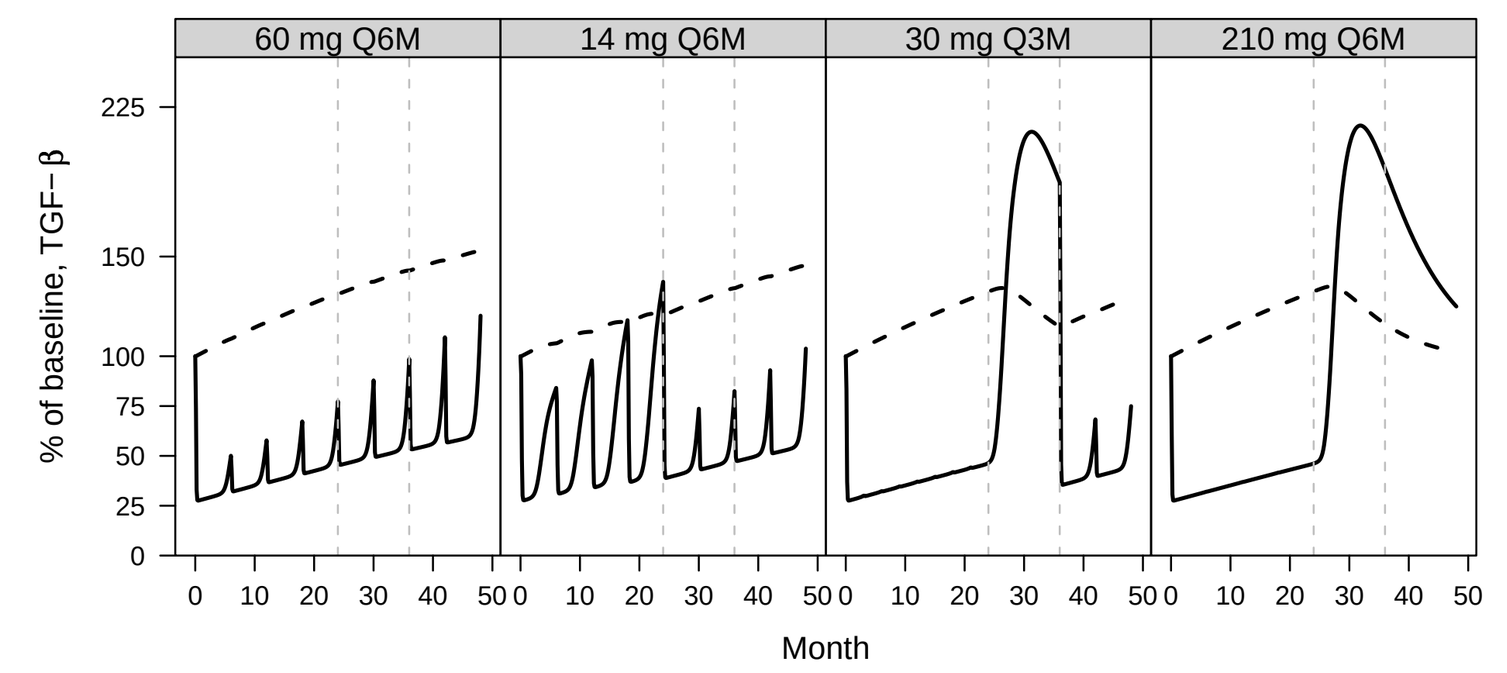
60 mg Q6M
14 mg Q6M
30 mg Q3M
210 mg Q6M
% of baseline, TGF−β
Month

Figure 9: *MSPM simulated active (solid lines) and latent (dashed lines) TGF-β after multiple denosumab doses. Figure taken from Peterson and Riggs[6]*

RESULTS – MSPM QUALIFICATION USING *FREEDOM* DATA

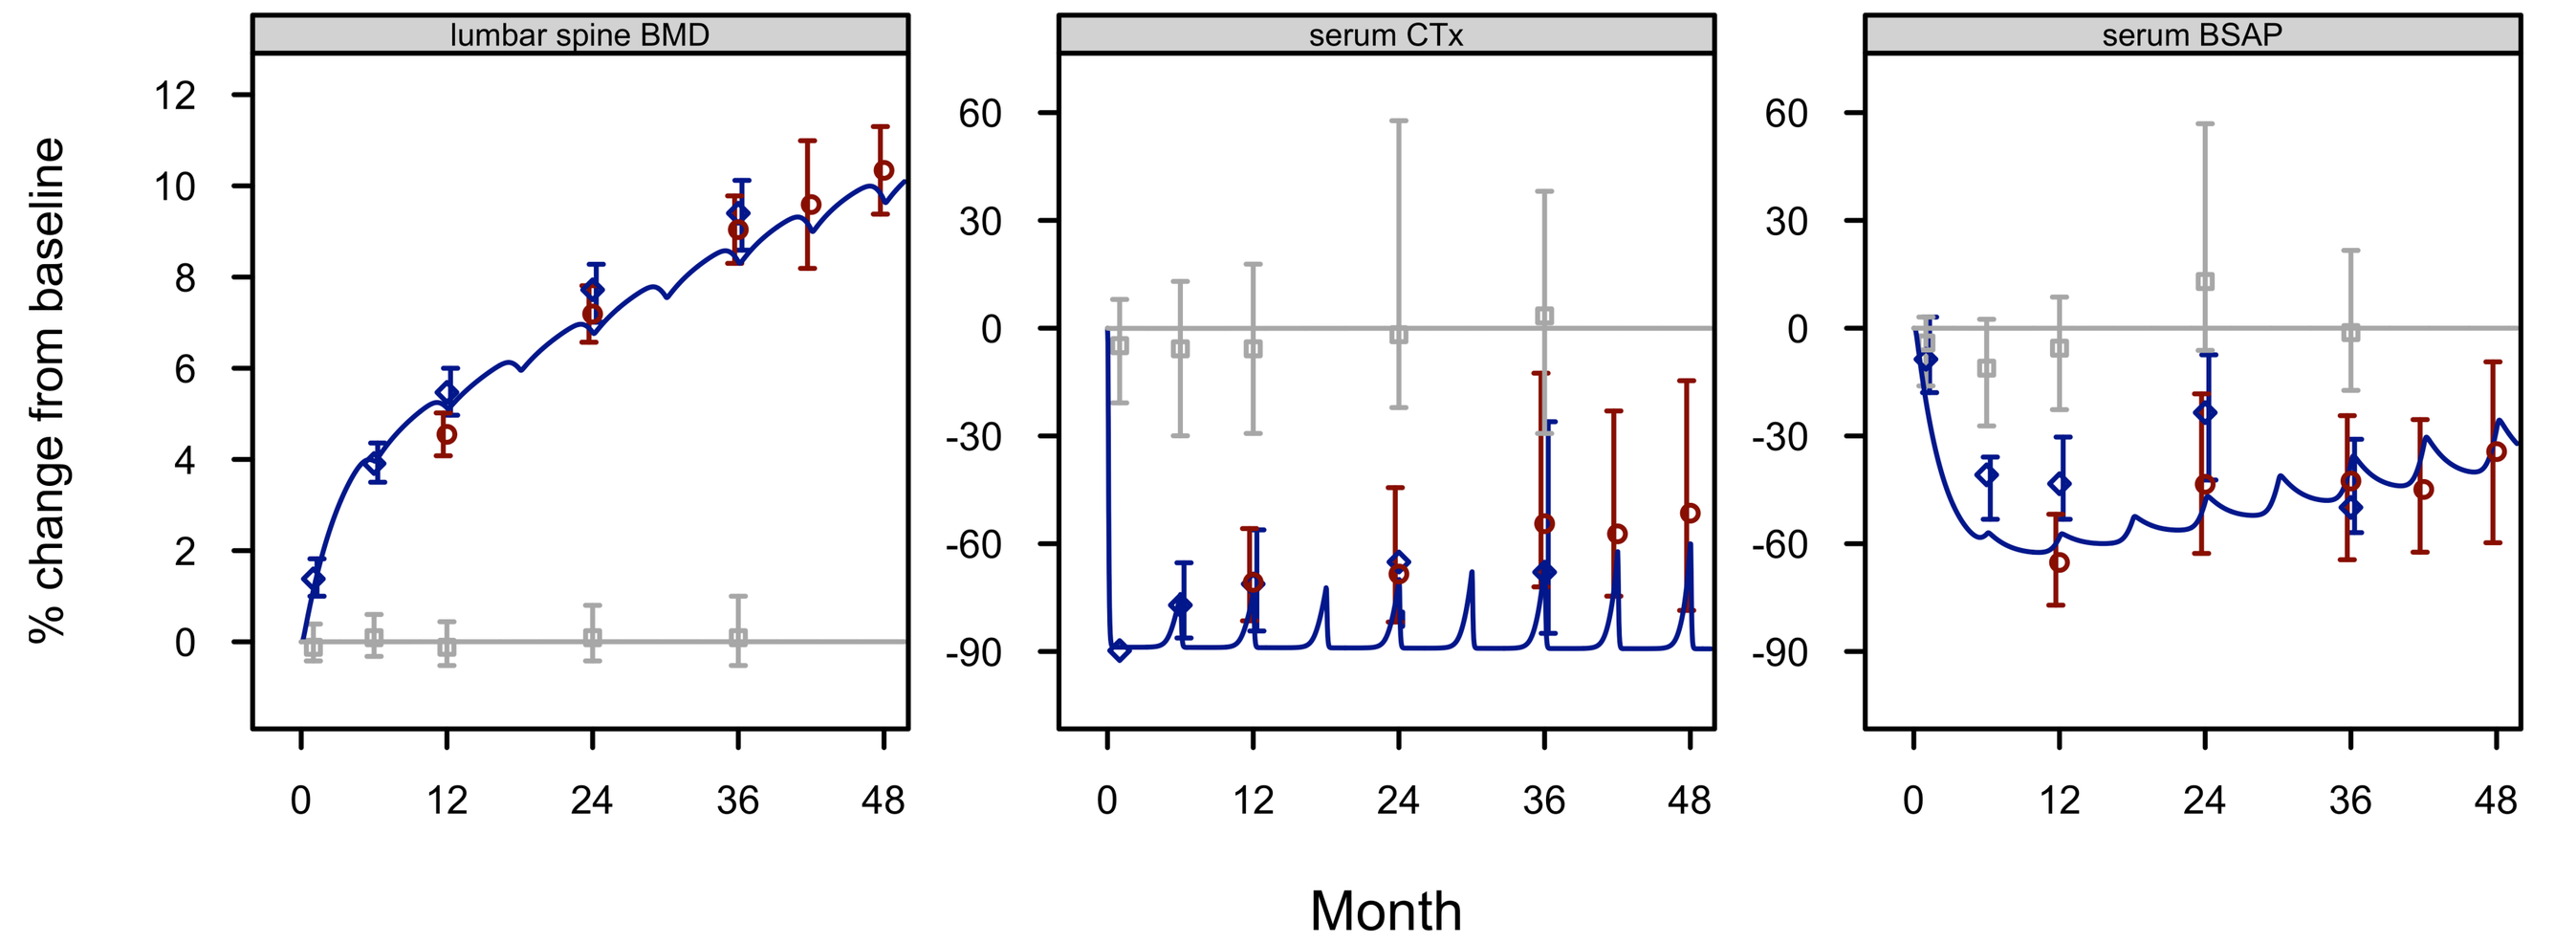
lumbar spine BMD
serum CTx
serum BSAP
% change from baseline
Month

Figure 10: *Observed (symbols) and simulated (lines) BMD, CTx, and BSAP during during treatment with 60mg Q6M denosumab for 4 years. Observed values from denosumab treatment groups: NCT00089791 (FREEDOM, blue symbols)[3, 4] and NCT00043186 (red symbols);[10, 12] and placebo treatment group: NCT00089791 (grey symbols).*

- MSPM predicted LS BMD changes: 1.1, 4.0, 5.2, 6.1, 6.9, 8.5, 9.2 and 9.8% at 1, 6, 12, 18, 24, 36, 42 and 48 months, respectively, following denosumab 60 mg Q6 months
- MSPM predictions were in close agreement with observed data: Mean absolute percentage error=9.1%; Mean percentage error=-7.9%
- MSPM also predicted the nearly complete decline in osteoclast function with slight increase in pre-dose (6 months after previous dose) BTMs with continued administration

CONCLUSIONS

- External data qualified the performance of the existing MSPM for prediction of BTM and BMD changes associated with denosumab treatment
- Indicated MSPM predictions can be generalized across data sets
- Provided further confidence in model-based predictions of physiologic changes due to RANKL modulation and in related decision-making in drug development and clinical practice

REFERENCES

[1] Peterson, M.C. and Riggs, M.M. A physiologically based mathematical model of integrated calcium homeostasis and bone remodeling. *Bone* **46** (2010):49–63.
[2] Riggs, M.M., Peterson, M.C. and Gastonguay, M.R. Multiscale physiology-based modeling of mineral bone disorder in patients with impaired kidney function. *J Clin Pharmacol* **52** (2012):45S–53S.
[3] Eastell, R., Christiansen, C., Grauer, A., Kutilek, S., Libanati, C., McClung, M.R., Reid, I.R., Resch, H., Siris, E., Uebelhart, D., Wang, A., Weryha, G. and Cummings, S.R. Effects of denosumab on bone turnover markers in postmenopausal osteoporosis. *J Bone Miner Res* **26** (2011):530–7.
[4] Cummings, S.R., San Martin, J., McClung, M.R., Siris, E.S., Eastell, R., Reid, I.R., Delmas, P., Zoog, H.B., Austin, M., Wang, A., Kutilek, S., Adami, S., Zanchetta, J., Libanati, C., Siddhanti, S., Christiansen, C. and FREEDOM Trial. Denosumab for prevention of fractures in postmenopausal women with osteoporosis. *N Engl J Med* **361** (2009):756–65.
[5] Riggs, M.M. Multiscale systems models as a knowledge bridge between biology, physiology, and pharmacology. *AAPS Newsmagazine* (2011).
[6] Peterson, M. and Riggs, M. Predicting nonlinear changes in bone mineral density over time using a multiscale systems pharmacology model. *CPT Pharmacometrics Syst. Pharmacol* (2012 (in press)).
[7] Riggs, M., Bennetts, M., van der Graaf, P. and Martin, S. Integrated pharmacometrics and systems pharmacology model-based analyses to guide gnrh receptor modulator development for management of endometriosis. *CPT Pharmacometrics Syst. Pharmacol* **1** (2012 (in press)).
[8] Plan, E.L., Baron, K.T., Gastonguay, M.R., French, J.L., Gillespie, W.R. and Riggs, M.M. Bayesian joint modeling of bone mineral density and repeated time-to-fracture event for multiscale bone systems model extension. In *PAGE 21st Meeting* (2012).
[9] R Development Core Team. *R: A Language and Environment for Statistical Computing*. R Foundation for Statistical Computing, Vienna, Austria (2008). ISBN: 3-900051-07-0. URL `http://www.R-project.org`
[10] McClung, M.R., Lewiecki, E.M., Cohen, S.B., Bolognese, M.A., Woodson, G.C., Moffett, A.H., Peacock, M., Miller, P.D., Lederman, S.N., Chesnut, C.H., Lain, D., Kivitz, A.J., Holloway, D.L., Zhang, C., Peterson, M.C., Bekker, P.J. and AMG 162 Bone Loss Study Group. Denosumab in postmenopausal women with low bone mineral density. *N Engl J Med* **354** (2006):821–31.
[11] Peterson, M., Stouch, B., Chen, D., Baughman, S., Holloway, D., Bekker, P. and Martin, S. A population pk/pd model describes the rapid, profound, and sustained suppression of urinary n-telopeptide following administration of amg 162, a fully human monoclonal antibody against rankl, to healthy postmenopausal women. *The AAPS Journal* **24** (2004).
[12] Miller, P.D., Bolognese, M.A., Lewiecki, E.M., McClung, M.R., Ding, B., Austin, M., Liu, Y. and San Martin, J. Effect of denosumab on bone density and turnover in postmenopausal women with low bone mass after long-term continued, discontinued, and restarting of therapy: a randomized blinded phase 2 clinical trial. *Bone* **43** (2008):222–229.

Abstract #SU0363: Presented at American Society of Bone Mineral Research (ASBMR) Annual Meeting; Minneapolis, MN; 14-October 2012

Copies available at: www.metrumrg.com/publications

© Metrum Research Group LLC 2012